SUN-257

Lorundrostat Modulates Heart Failure Risk Biomarkers in Participants With Uncontrolled Hypertension

Michael A. Tortorici, Kushan L. Gunawardhana, Nicolas De Jay, and David M. Rodman

Mineralys Therapeutics, Radnor, PA

MINERALYS

BACKGROUND

- Heart failure (HF) with preserved ejection fraction (HFpEF) is characterized by impaired relaxation, cardiometabolic disease, and often involves maladaptive activation of the renin-angiotensin-aldosterone system (RAAS)[1]
- Independent predictors of incident HFpEF include older age, higher systolic blood pressure (SBP), increased body-mass index, diabetes, antihypertensive treatment, atrial fibrillation, and previous myocardial infarction
- Lorundrostat is a novel aldosterone synthase inhibitor with demonstrated efficacy in reducing SBP in participants with uncontrolled hypertension, including treatment-resistant hypertension, in Phase 2b and Phase 3 trials[2,3]

OBJECTIVE

- To conduct a hypothesis-generating analysis to characterize the systemic pharmacodynamic (PD) effects and potential modulation of pathways implicated in HF pathophysiology with lorundrostat treatment in participants with uncontrolled hypertension, including treatment-resistant hypertension

METHODS

- Serum samples were collected at baseline and at 12 weeks from participants randomized to lorundrostat (n=733) or placebo (n=271) in the Phase 3 Launch-HTN[3] and Phase 2b Advance-HTN[2] (**Table 1**)
- Circulating biomarkers were measured using a custom 1053 aptamer panel based on the SomaLogic cardiovascular protein panel (SomaScan) and literature-curated targets
- Biomarker data underwent quality control (QC) to remove low-quality samples and aptamers, normalize for batch/plate effects, and account for relevant technical and biological covariates (**Figure 1**)
- Placebo-adjusted treatment effects were estimated using linear mixed models within each trial, adjusting for key demographic and technical covariates, and meta-analyzed across trials using inverse-variance fixed effects
- To evaluate whether lorundrostat reduces coordinated sets of risk-associated biomarkers, we performed gene set enrichment analysis (GSEA) using published biomarker sets
- Finally, we mapped modules with aggregated, coordinated longitudinal changes across related biomarkers to identify hub biomarkers within each module using weighted gene co-expression network analysis (WGCNA)[4]

Table 1. Serum Samples Were Collected From 1004 Participants From Large Hypertension Trials

Study	Placebo Participants	Lorundrostat Participants	Total Participants
Launch-HTN *MLS-101-301 (Ph 3)*	188	574	762
Advance-HTN *MLS-101-202 (Ph 2)*	83	159	242
Total	271	733	1004

Launch-HTN and Advance-HTN were double-blind, placebo-controlled trials that evaluated the effect of lorundrostat on systolic blood pressure.

Ph, phase.

RESULTS

Figure 1. Comprehensive Quality Control Reveals High-Quality Biomarker Data

1 **97.2% of samples pass QC**
- Verified balanced baseline covariates
- Flagged technical outliers

2 **90.1% of aptamers pass QC**
- Profiled ~1.8% duplicate samples to identify low-reproducibility aptamers
- Flagged low-quality technical outliers

3 **Data Processing**
- Normalized for batch and plate effects
- Modeled technical (plate, site) and biological covariates

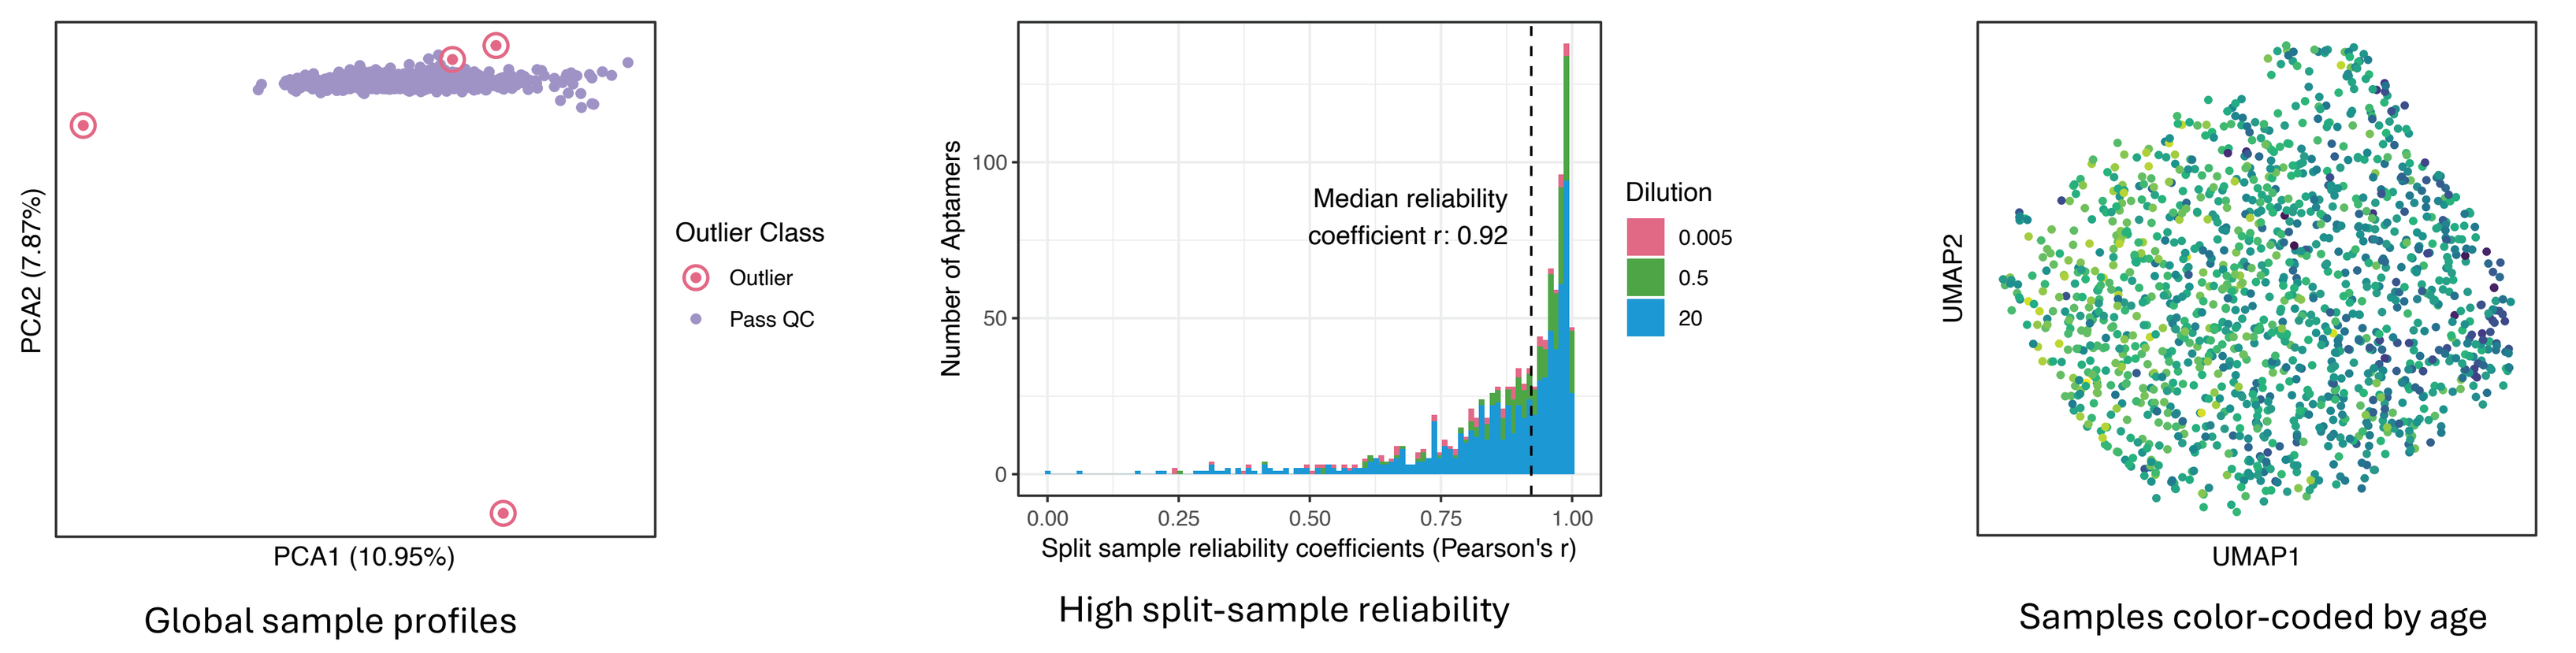
PCA2 (7.87%)
PCA1 (10.95%)
Outlier Class
Outlier
Pass QC
Global sample profiles
Number of Aptamers
Median reliability coefficient r: 0.92
Dilution
0.005
0.5
20
Split sample reliability coefficients (Pearson's r)
High split-sample reliability
UMAP2
UMAP1
AGE
Samples color-coded by age

Data shown are from Launch-HTN; Advance-HTN yielded comparable results.

PCA, principal component analysis; UMAP, Uniform Manifold Approximation and Projection.

Figure 2. Lorundrostat Demonstrates Robust Systemic PD Effects and RAAS Target Engagement

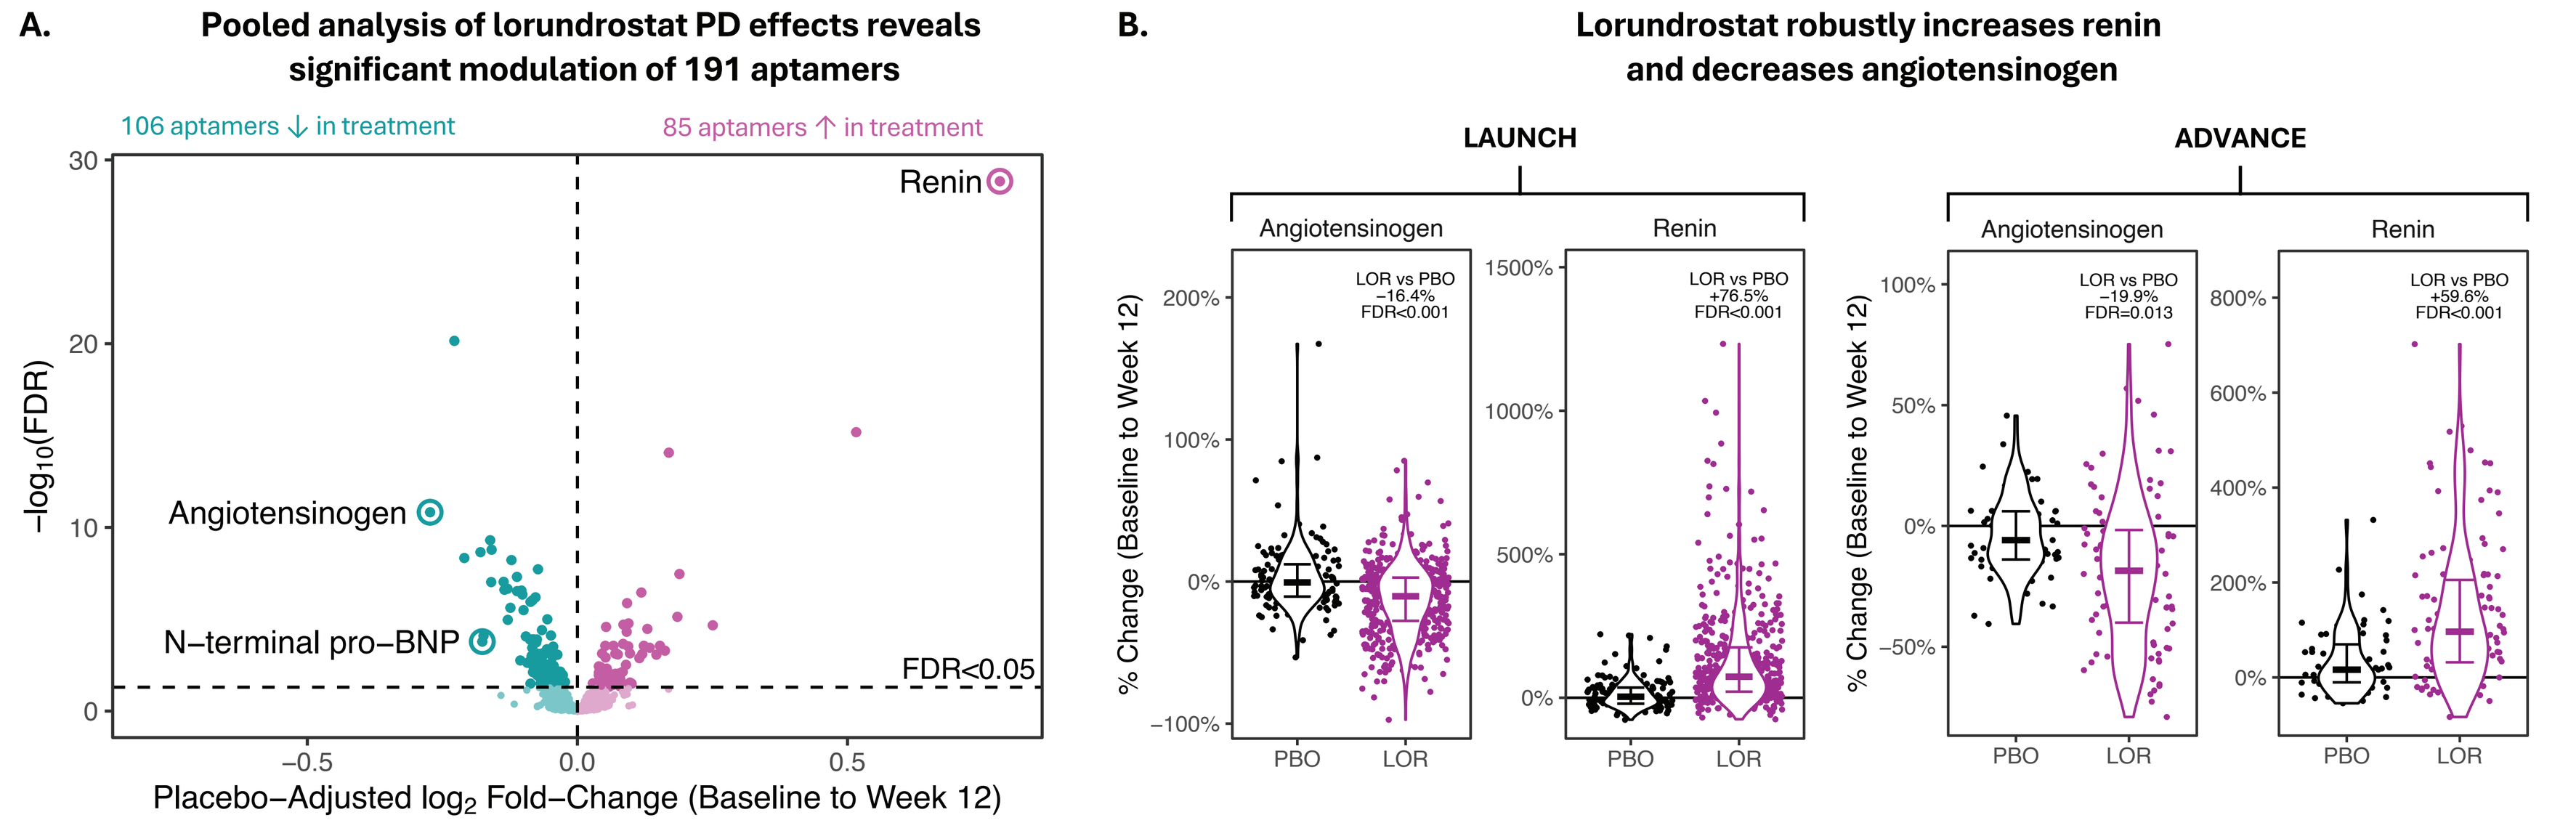
A. Pooled analysis of lorundrostat PD effects reveals significant modulation of 191 aptamers
106 aptamers ↓ in treatment
85 aptamers ↑ in treatment
Renin
Angiotensinogen
N–terminal pro–BNP
FDR<0.05
−log10(FDR)
Placebo–Adjusted log2 Fold–Change (Baseline to Week 12)
B. Lorundrostat robustly increases renin and decreases angiotensinogen
LAUNCH
ADVANCE
Angiotensinogen
Renin
LOR vs PBO −16.4% FDR<0.001
LOR vs PBO +76.5% FDR<0.001
LOR vs PBO −19.9% FDR=0.013
LOR vs PBO +59.6% FDR<0.001
% Change (Baseline to Week 12)
PBO
LOR

BNP, B-type natriuretic peptide; FDR, false discovery rate; LOR, lorundrostat; PBO, placebo; PD, pharmacodynamic.

- Lorundrostat treatment significantly decreased the expression of 106 aptamers and increased the expression of 85 aptamers vs placebo (false discovery rate [FDR] < 0.05; **Figure 2A**)
- Robust increases in renin (placebo-adjusted change, 59.6% to 76.5%) and decreases in angiotensinogen (−19.9% to −16.4%) reflect RAAS target engagement (**Figure 2B**)

Figure 3. Lorundrostat Decreases Biomarkers Associated With Incident Heart Failure Risk at 12 Weeks

A. Incident heart failure risk biomarker sets compiled from published literature

Study	Cohorts	MR	Number of Biomarkers	Assay
Shah et al (2024)[5]	**ARIC; HUNT**	**√**	**11**	**Soma**
Shah et al (2024)[5]	ARIC; HUNT	X	17	Soma
Lind et al (2021)[6]	PIVUS; ULSAM	X	35	Olink

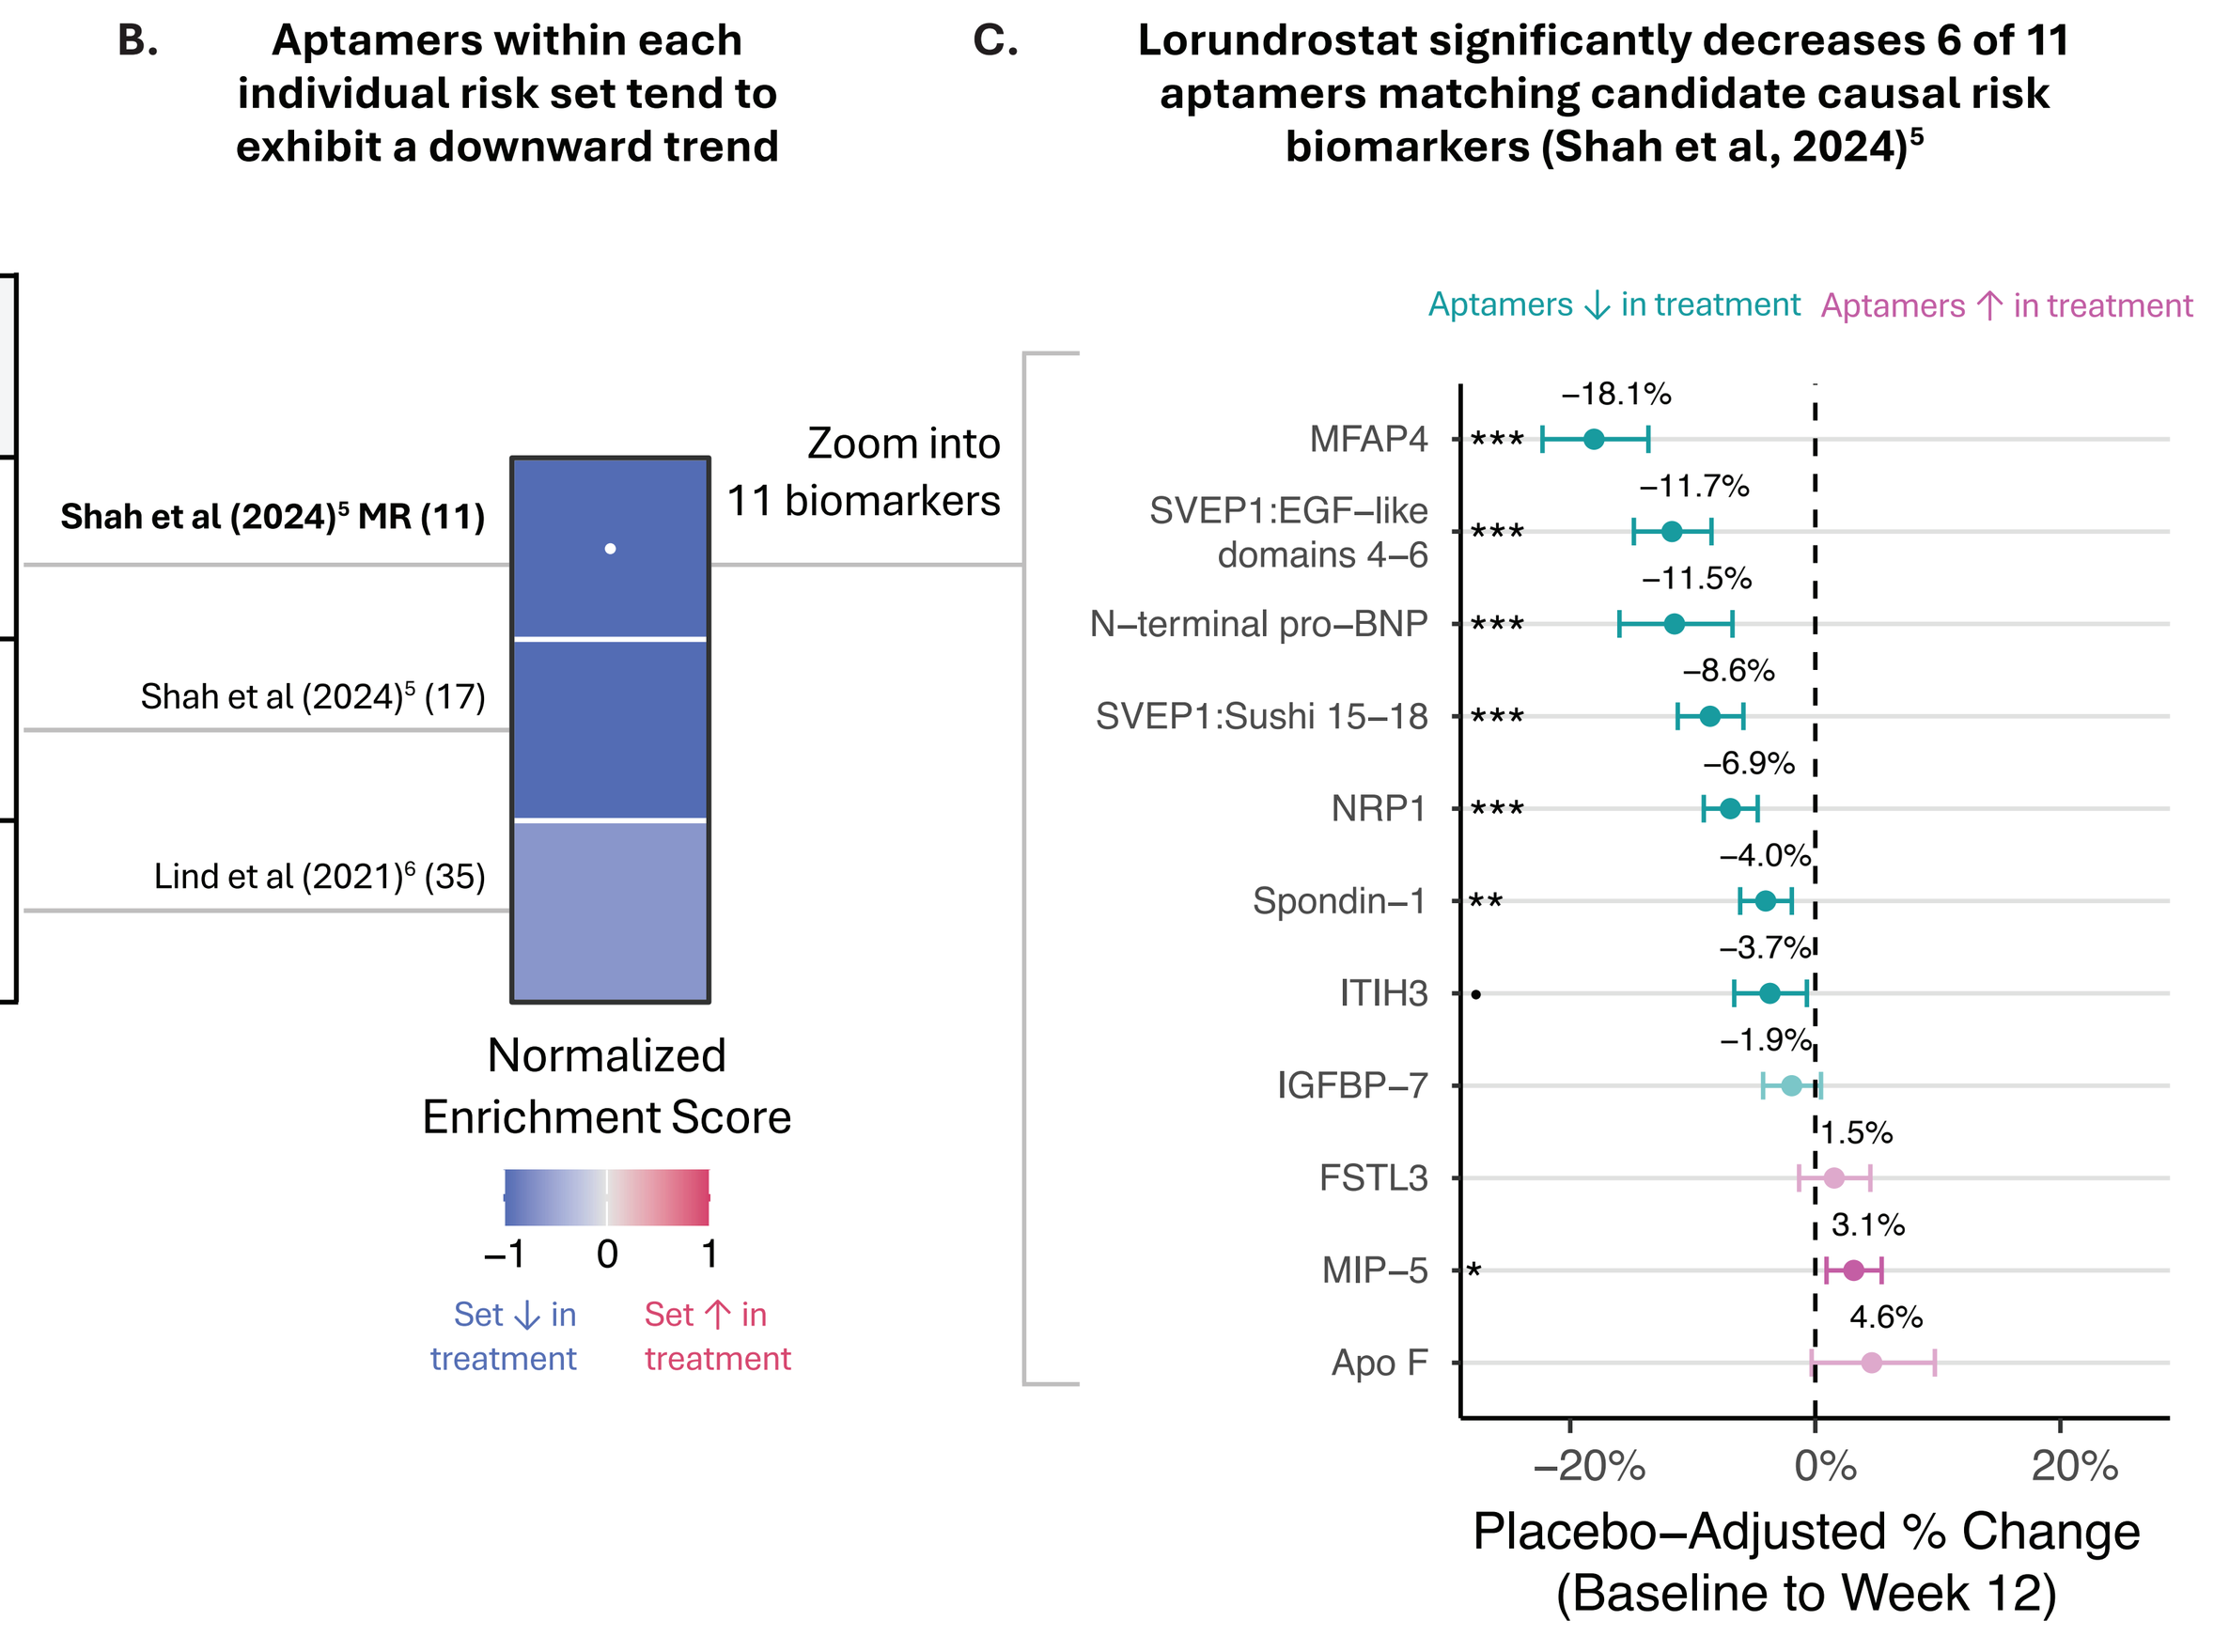
B. Aptamers within each individual risk set tend to exhibit a downward trend
Shah et al (2024)5 MR (11)
Shah et al (2024)5 (17)
Lind et al (2021)6 (35)
Zoom into 11 biomarkers
Normalized Enrichment Score
−1 0 1
Set ↓ in treatment
Set ↑ in treatment
C. Lorundrostat significantly decreases 6 of 11 aptamers matching candidate causal risk biomarkers (Shah et al, 2024)5
Aptamers ↓ in treatment
Aptamers ↑ in treatment
MFAP4 *** −18.1%
SVEP1:EGF–like domains 4–6 *** −11.7%
N–terminal pro–BNP *** −11.5%
SVEP1:Sushi 15–18 *** −8.6%
NRP1 *** −6.9%
Spondin–1 ** −4.0%
ITIH3 • −3.7%
IGFBP–7 −1.9%
FSTL3 1.5%
MIP–5 * 3.1%
Apo F 4.6%
−20% 0% 20%
Placebo–Adjusted % Change (Baseline to Week 12)

•FDR<0.10; *FDR<0.05; **FDR<0.01; ***FDR<0.001.

APOF, apolipoprotein F; BNP, B-type natriuretic peptide; FDR, false discovery rate; FSTL3, follistatin-like protein 3; HF, heart failure; IGFBP-7, insulin-like growth factor-binding protein 7; ITIH3, inter-alpha-trypsin inhibitor heavy chain H3; MFAP4, microfibrillar-associated protein 4; MIP-5, macrophage inflammatory protein 5; MR, Mendelian randomization; NRP1, neuropilin-1; PD, pharmacodynamic; SVEP1, Sushi, von Willebrand factor type A, EGF, and pentraxin domain-containing protein 1.

- Within each set of incident HF risk biomarkers compiled from published literature,[5,6] corresponding aptamers exhibit a collective downward trend following 12 weeks of lorundrostat treatment (**Figures 3A** and **3B**)
- Lorundrostat significantly decreases 6 out of 11 aptamers previously identified as candidate causal risk biomarkers,[5] consistent with the hypothesis that RAAS inhibition favorably modulates processes involved in HF risk (**Figure 3C**)

Figure 4. Lorundrostat Is Associated With a Coordinated Reduction in HF Risk, Fibrosis, and Matrix Remodeling Biomarkers

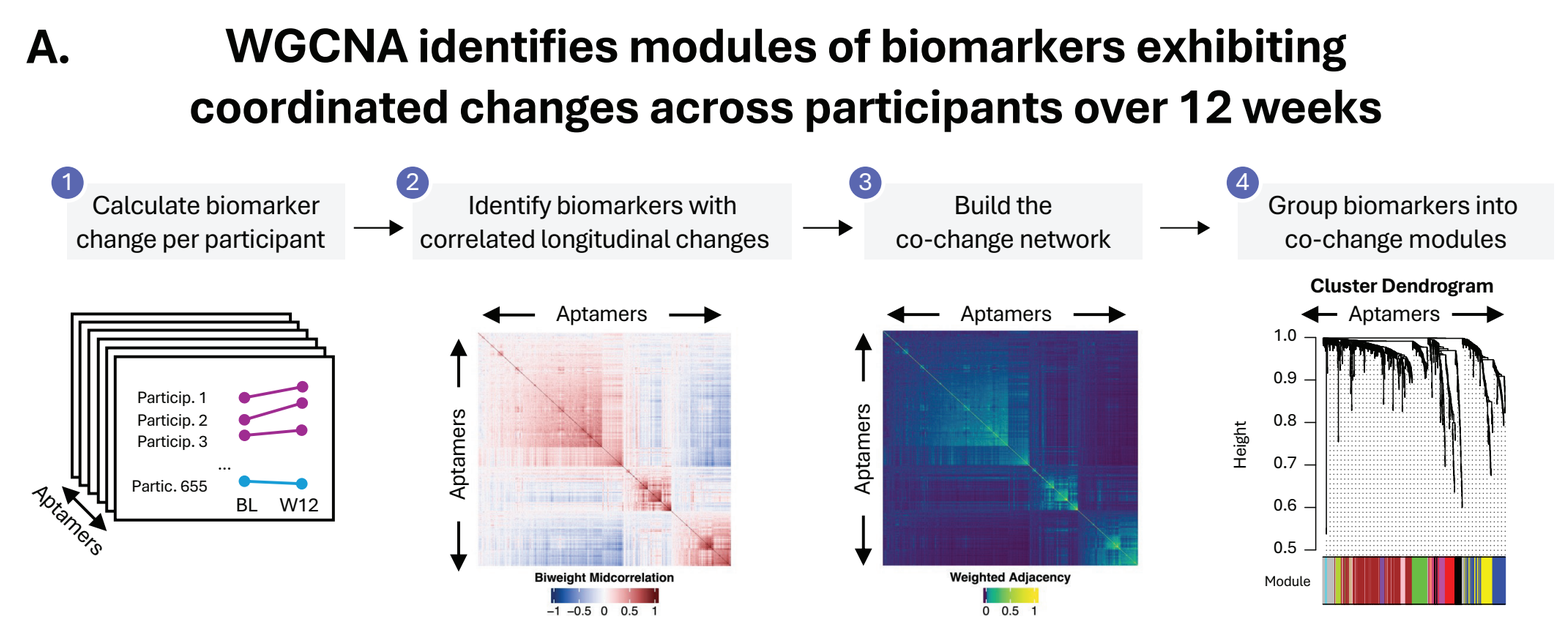
A. WGCNA identifies modules of biomarkers exhibiting coordinated changes across participants over 12 weeks
1 Calculate biomarker change per participant
2 Identify biomarkers with correlated longitudinal changes
3 Build the co-change network
4 Group biomarkers into co-change modules
Aptamers
Cluster Dendrogram
Biweight Midcorrelation
Weighted Adjacency

B. Lorundrostat significantly modulates 4 co-change modules

	Modules	Placebo-Adjusted Treatment Effect		Change in SBP in Treated Participants	
Module	# aptamers	β	FDR	β	FDR
pink	**45**	**-0.023**	***	**-0.005**	***
green	**75**	**-0.010**	*	-0.001	ns
brown	253	-0.008	·	-0.001	ns
red	57	-0.002	ns	+0.002	ns
green-yellow	22	-0.002	ns	-0.000	ns
salmon	12	0.000	ns	-0.002	ns
black	45	0.002	ns	-0.001	ns
magenta	41	0.002	ns	+0.001	ns
grey	64	0.004	ns	-0.002	ns
cyan	10	0.006	ns	+0.001	ns
purple	22	0.006	ns	+0.005	***
blue	110	0.008	·	-0.001	ns
yellow	**79**	**0.014**	***	+0.001	ns
tan	**15**	**0.018**	***	**+0.008**	***

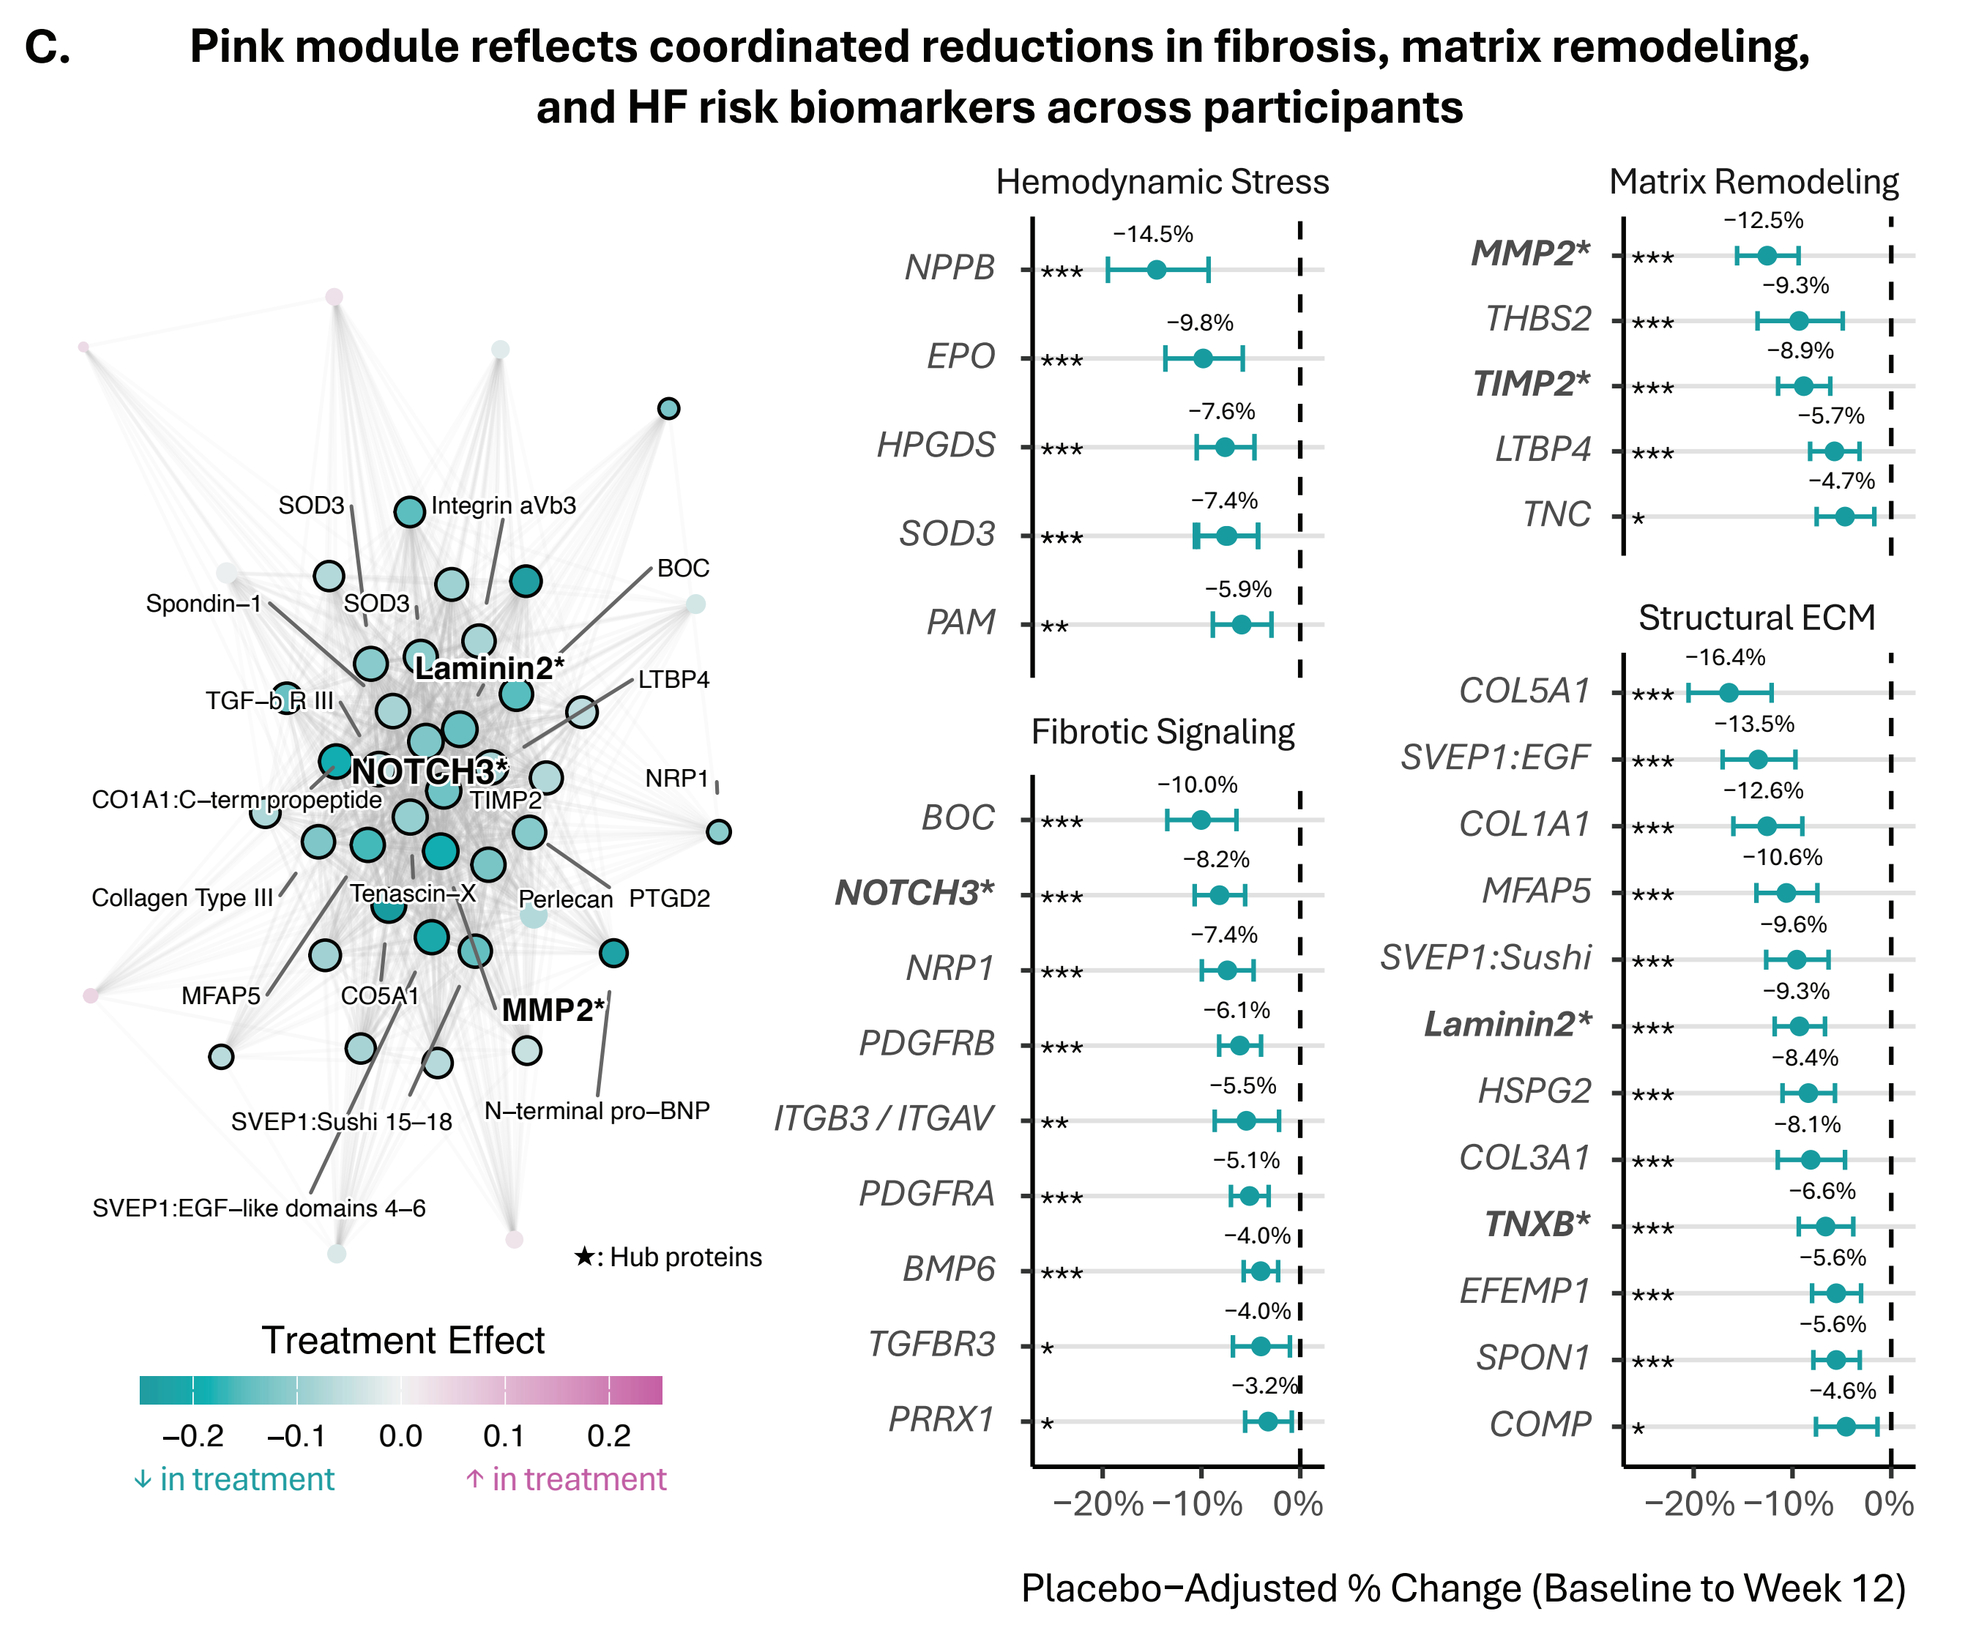
C. Pink module reflects coordinated reductions in fibrosis, matrix remodeling, and HF risk biomarkers across participants
Laminin2*
NOTCH3*
MMP2*
★: Hub proteins
Treatment Effect
↓ in treatment
↑ in treatment
Hemodynamic Stress
NPPB *** −14.5%
EPO *** −9.8%
HPGDS *** −7.6%
SOD3 *** −7.4%
PAM ** −5.9%
Fibrotic Signaling
BOC *** −10.0%
NOTCH3* *** −8.2%
NRP1 *** −7.4%
PDGFRB *** −6.1%
ITGB3 / ITGAV ** −5.5%
PDGFRA *** −5.1%
BMP6 *** −4.0%
TGFBR3 * −4.0%
PRRX1 * −3.2%
Matrix Remodeling
MMP2* *** −12.5%
THBS2 *** −9.3%
TIMP2* *** −8.9%
LTBP4 *** −5.7%
TNC * −4.7%
Structural ECM
COL5A1 *** −16.4%
SVEP1:EGF *** −13.5%
COL1A1 *** −12.6%
MFAP5 *** −10.6%
SVEP1:Sushi *** −9.6%
Laminin2* *** −9.3%
HSPG2 *** −8.4%
COL3A1 *** −8.1%
TNXB* *** −6.6%
EFEMP1 *** −5.6%
SPON1 *** −5.6%
COMP * −4.6%
Placebo–Adjusted % Change (Baseline to Week 12)

•FDR<0.10; *FDR<0.05; **FDR<0.01; ***FDR<0.001.

ECM, extracellular matrix; HF, heart failure; MR, Mendelian randomization; ssGSEA, single-sample gene set enrichment analysis; WGCNA, weighted gene co-expression network analysis.

- In Launch-HTN, we identified 14 biomarker modules with coordinated longitudinal changes, 4 of which were significantly modulated by lorundrostat across participants (**Figures 4A** and **4B**)
- In particular, the pink module is defined by a hub of matrix-remodeling/fibrotic proteins, including laminin2, NOTCH3, and MMP-2 that decreased with treatment alongside structural extracellular matrix (ECM) proteins (eg, collagen-1/3/5) and MR-supported incident HF risk biomarkers such as NT-proBNP, SVEP1, NRP1, and spondin-1 (**Figure 4C**)
- The correlated reduction of these biomarkers within the same participants points to a broad and coherent pathway-level effect, consistent with the attenuation of fibrotic pathways, rather than isolated biomarker changes

Figure 5. Interparticipant Heterogeneity Suggests Distinct Molecular Treatment Trajectories

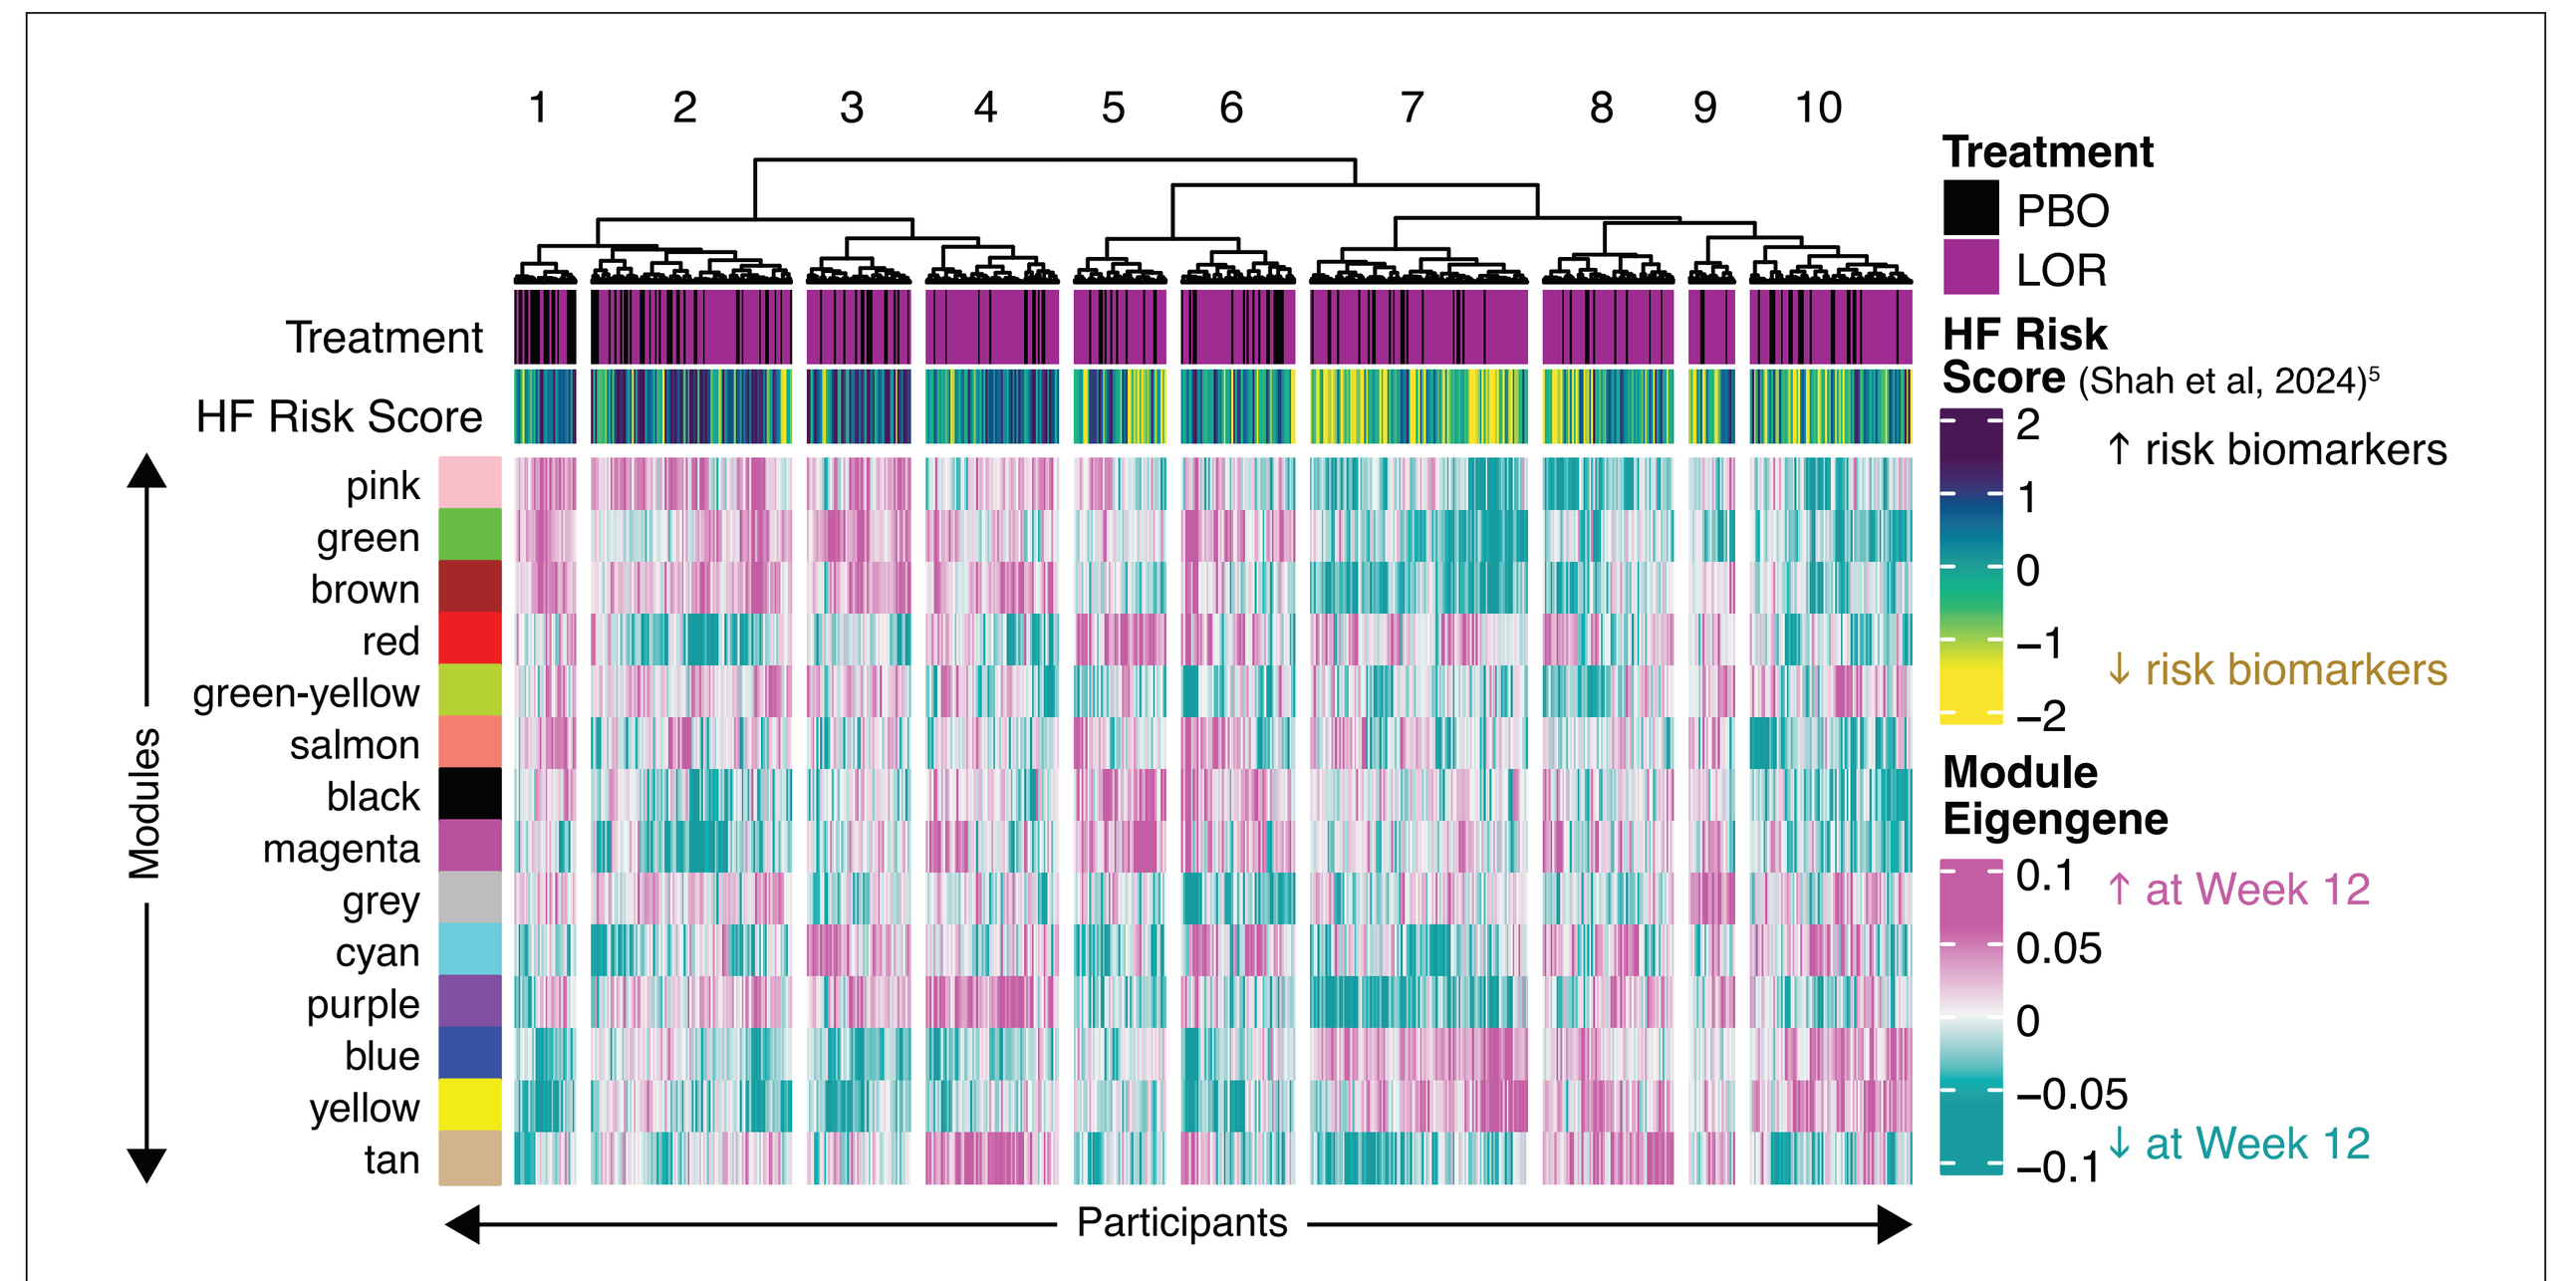
1 2 3 4 5 6 7 8 9 10
Treatment
HF Risk Score
Modules
pink
green
brown
red
green-yellow
salmon
black
magenta
grey
cyan
purple
blue
yellow
tan
Participants
Treatment
PBO
LOR
HF Risk Score (Shah et al, 2024)5
↑ risk biomarkers
↓ risk biomarkers
Module Eigengene
↑ at Week 12
↓ at Week 12

- Preliminary clustering reveals substantial interparticipant heterogeneity in longitudinal module response, suggesting distinct molecular trajectories warranting further investigation

CONCLUSIONS

- **Pharmacodynamic analysis confirmed significant increases in renin and decreases in angiotensinogen in treated participants, reflecting target engagement of the RAAS**
- **Lorundrostat was associated with significant reductions in 6 of 11 recently published candidate causal risk biomarkers of incident heart failure (eg, NT-proBNP),[5] consistent with the hypothesis that RAAS inhibition favorably modulates processes involved in HF risk**
- **Network analysis identified a co-change module with coordinated decreases in incident heart failure risk and ECM-related biomarkers under treatment. Their correlated reduction within the same participants points to a broad and coherent pathway-level effect, consistent with the attenuation of fibrotic pathways**
- **Although based on circulating biomarkers rather than clinical heart failure outcomes, these results provide biological plausibility and support further evaluation of the therapeutic potential of lorundrostat in HFpEF**

REFERENCES

1. Hamo CE, et al. *Nat Rev Dis Primers*. 2024;10(1):55.
2. Laffin LJ, et al. *N Engl J Med.* 2025;392(18):1813-23.
3. Saxena M, et al. *JAMA*. 2025;334(5):409-18.
4. Langfelder P, Horvath S. *BMC Bioinformatics*. 2008;9:559.
5. Shah AM, et al. *Nat Commun*. 2024;15(1):528.
6. Lind L, et al. *J Am Heart Assoc*. 2021;10(23):e023330.

ACKNOWLEDGMENTS

Advance-HTN and Launch-HTN were supported by Mineralys Therapeutics, Radnor, PA. Medical writing and editorial assistance were provided by John Watson, PhD, and Stephen Bublitz, ELS, of the Sensified Division of Woven Health Collective, LLC (New York, NY, USA), and were funded by Mineralys Therapeutics.

DISCLOSURES

MAT, KLG, NDJ, DMR: Employment with Mineralys Therapeutics and may have an equity interest in the company.

Presented at ENDO 2026, June 13-16, 2026, Chicago, IL