TH-PO1121

Identification of a Hypertensive Endotype with a Median Treatment Effect of -32 mmHg in Response to the Novel Aldosterone Synthase Inhibitor Lorundrostat

Borut Cizman[1], Luke Laffin[2], Natasa Rajicic[3], and David M. Rodman[1]

[1]Mineralys Therapeutics, Radnor, PA; [2]Cleveland Clinic Center for Clinical Research, Cleveland, OH; [3]Cytel Inc, Waltham, MA

INTRODUCTION

- Effective, precision directed treatment of patients with uncontrolled and treatment-resistant hypertension who do not respond to current standard-of-care antihypertensive therapy is an urgent unmet need
- Dysregulated aldosterone is growing in prevalence, contributes to uncontrolled hypertension (uHTN), and may be particularly important in individuals with obesity[1]
- Lorundrostat, a novel, highly selective aldosterone synthase inhibitor (ASI), led to substantial blood pressure (BP) reduction in adults with uHTN and an enhanced response in the subset of individuals with obesity in the TARGET-HTN trial[2]

Figure 1. Hypertensive endotype

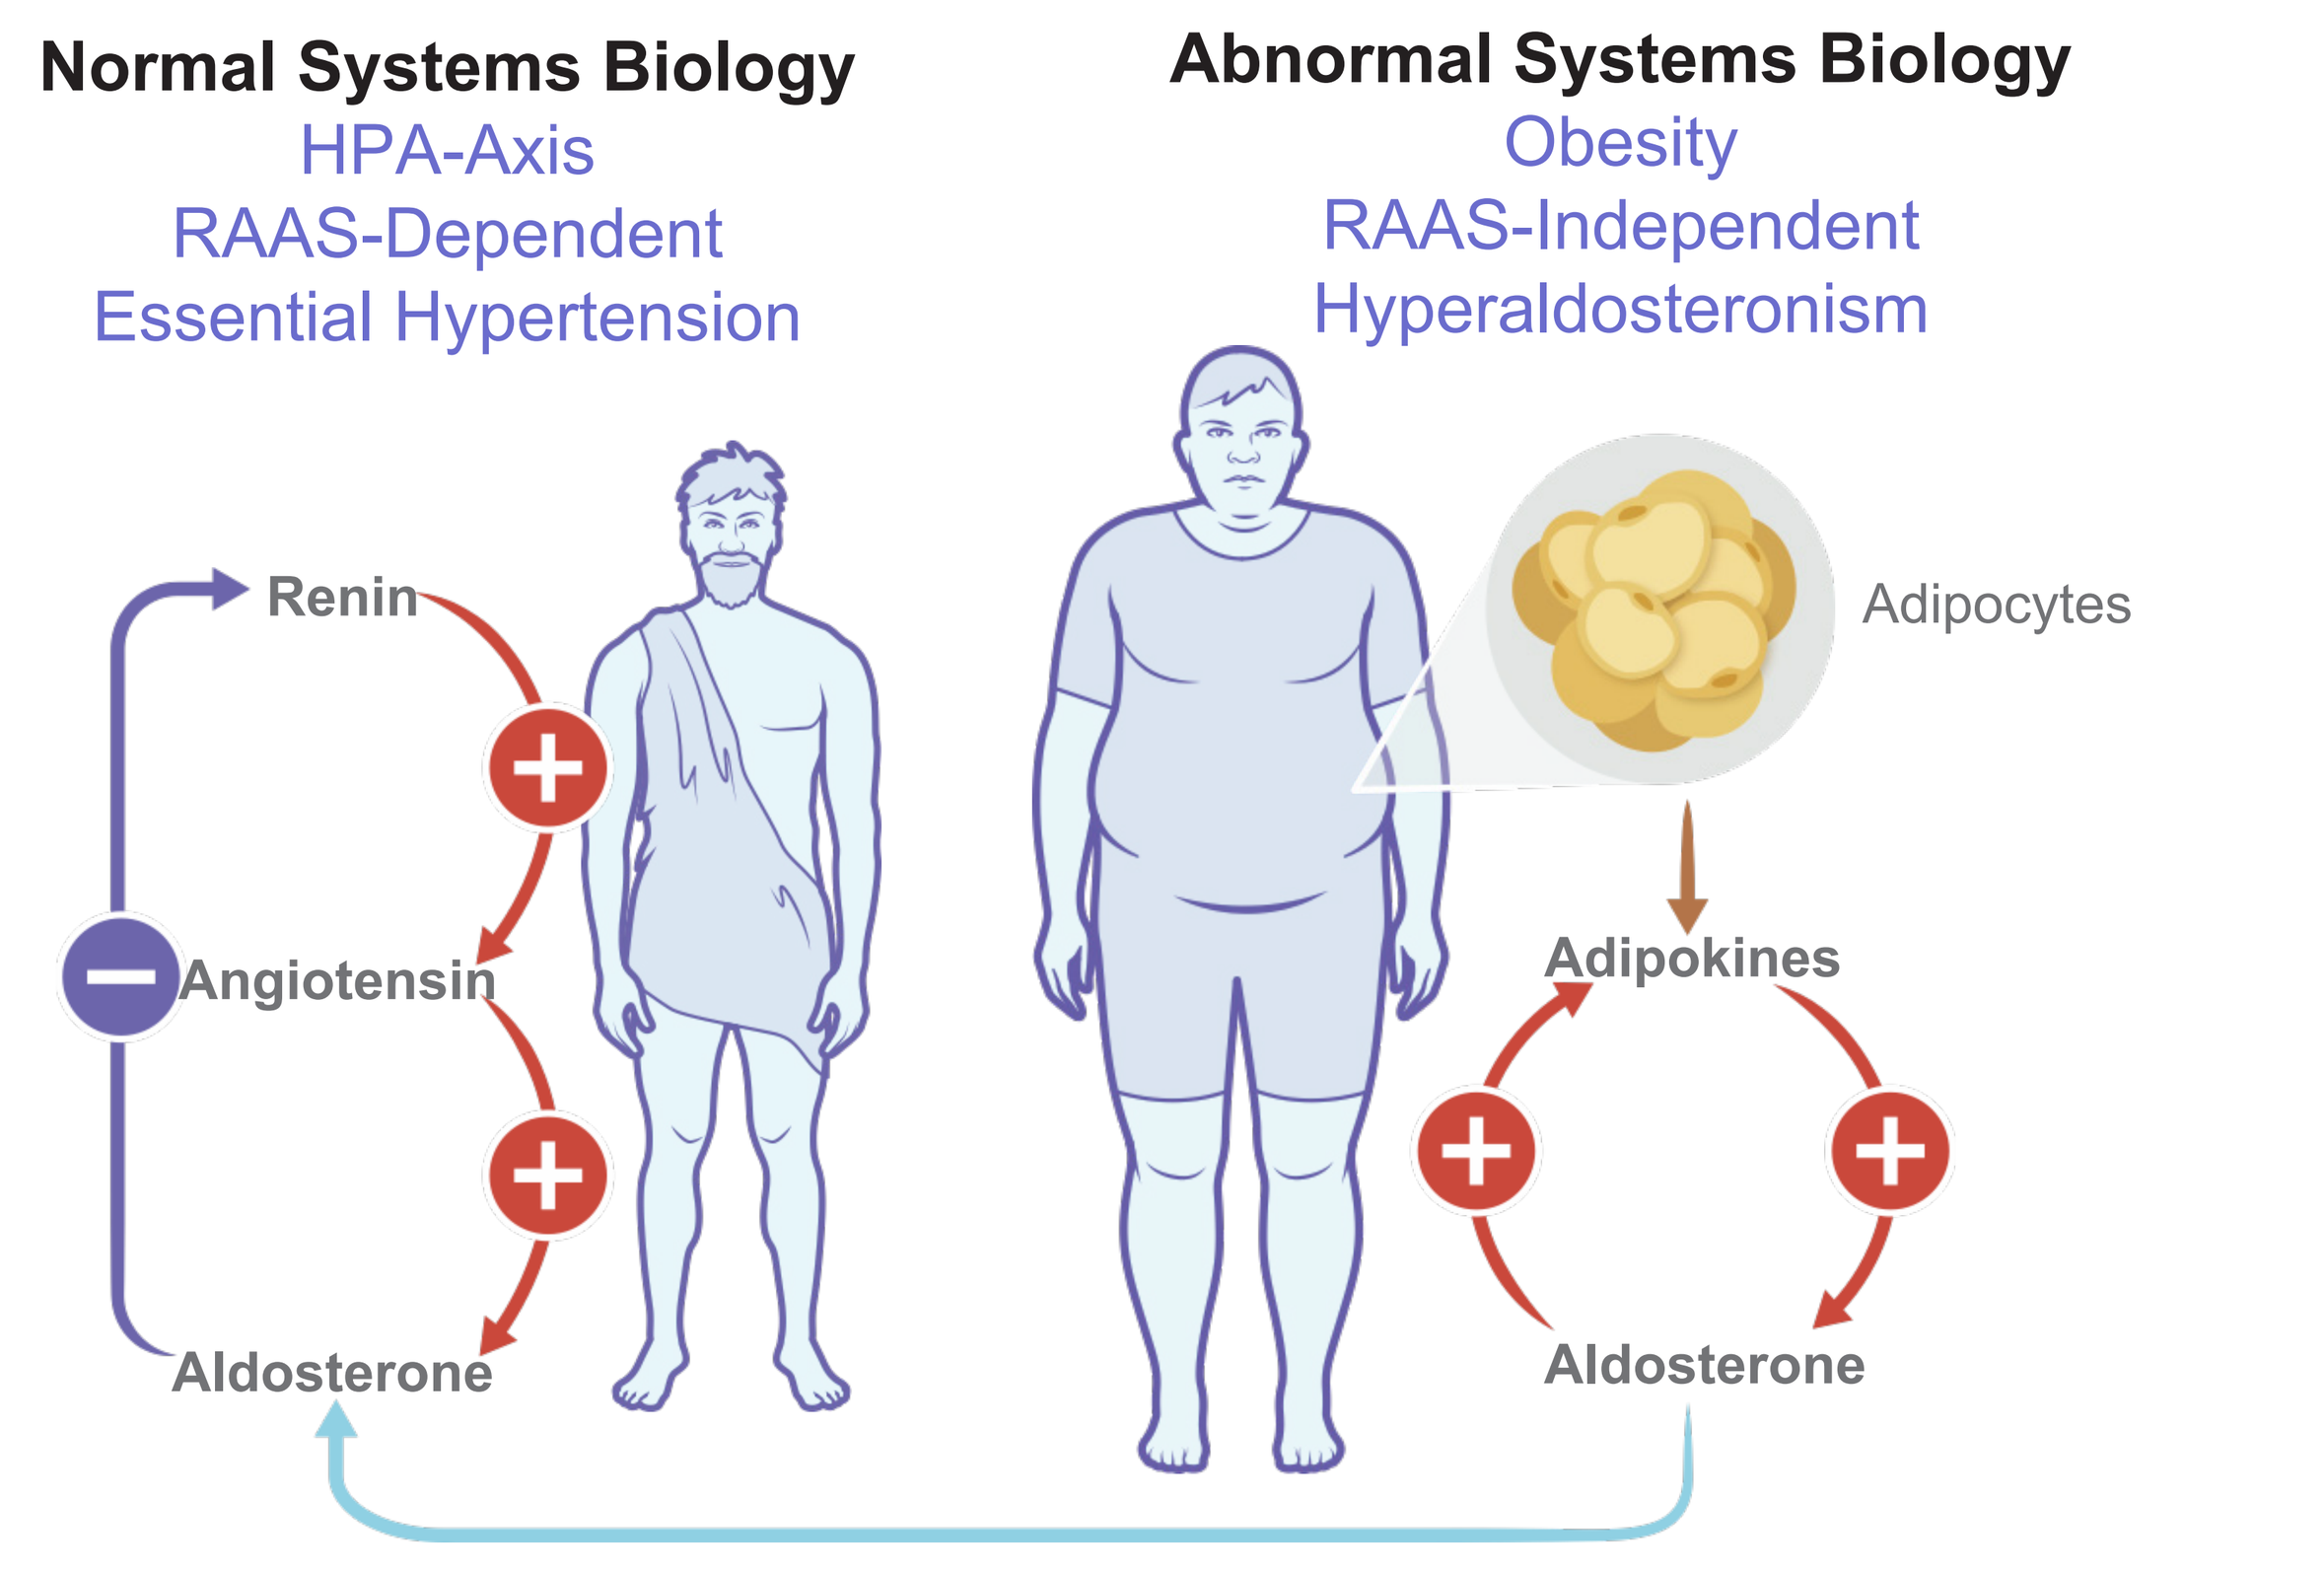
Normal Systems Biology
HPA-Axis
RAAS-Dependent
Essential Hypertension
Abnormal Systems Biology
Obesity
RAAS-Independent
Hyperaldosteronism
Renin
Angiotensin
Aldosterone
Adipocytes
Adipokines
Aldosterone

HPA, hypothalamus-pituitary-adrenal; RAAS, renin-angiotensin-aldosterone system.

OBJECTIVES

- To identify the adults with uHTN despite taking ≥2 antihypertensives who had an enhanced systolic BP reduction with lorundrostat treatment in TARGET-HTN
- To characterize the hypertensive endotype that supports an aldosterone targeted, precision directed treatment in uHTN

METHODS

Figure 2. TARGET-HTN study design

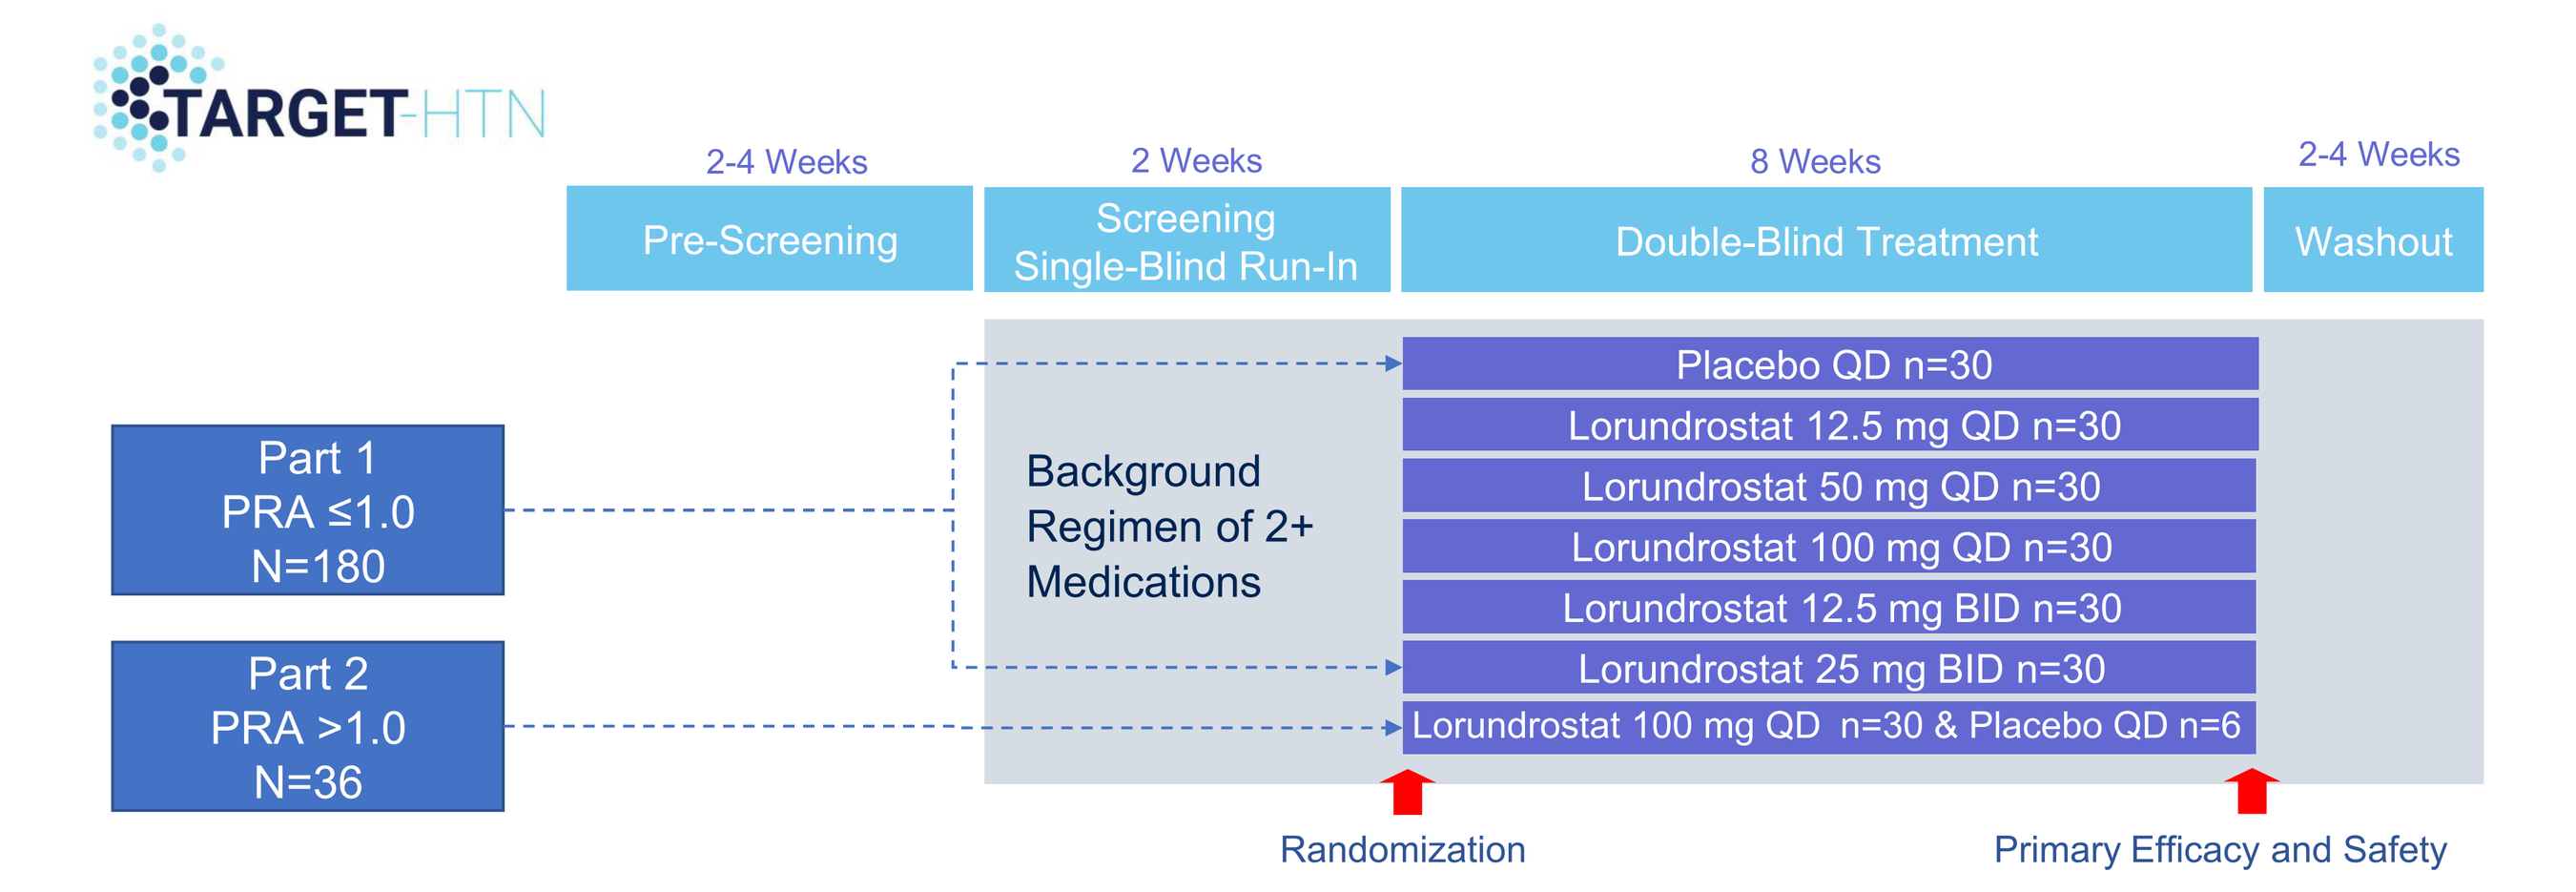
TARGET-HTN
2-4 Weeks
Pre-Screening
2 Weeks
Screening
Single-Blind Run-In
8 Weeks
Double-Blind Treatment
2-4 Weeks
Washout
Part 1
PRA ≤1.0
N=180
Part 2
PRA >1.0
N=36
Background
Regimen of 2+
Medications
Placebo QD n=30
Lorundrostat 12.5 mg QD n=30
Lorundrostat 50 mg QD n=30
Lorundrostat 100 mg QD n=30
Lorundrostat 12.5 mg BID n=30
Lorundrostat 25 mg BID n=30
Lorundrostat 100 mg QD n=30 & Placebo QD n=6
Randomization
Primary Efficacy and Safety

BID, twice daily; PRA, plasma renin activity; QD, once daily.

Table 1. Baseline demographics of non-obese and obese subjects in TARGET-HTN

Full Analysis Set (N=200), n (%)	BMI 25 to 29.9 (n=71)	BMI ≥30 (n=107)
Using 2 baseline HT meds	43 (60.6)	56 (52.3)
Using 3+ baseline HT meds	28 (39.4)	51 (47.7)
Using thiazide at baseline	37 (52.1)	66 (61.7)
Baseline systolic BP by AOBP <140 mmHg	35 (49.3)	52 (48.6)
Baseline systolic BP by AOBP ≥140 mmHg	36 (50.7)	55 (51.4)
African American	23 (32.4)	46 (43.0)
Male	30 (42.3)	43 (40.2)
AOBP systolic BP (mean, SD)	141.5 (9.6)	143 (10.9)
ABPM 24h systolic BP (mean, SD)	135.0 (13.2)	140.1 (14.0)

ABPM, ambulatory blood pressure monitoring; AOBP, automated office blood pressure; BMI, body mass index; BP, blood pressure; HT, hypertension; SD, standard deviation.

Figure 3. Percent change in serum aldosterone at week 4

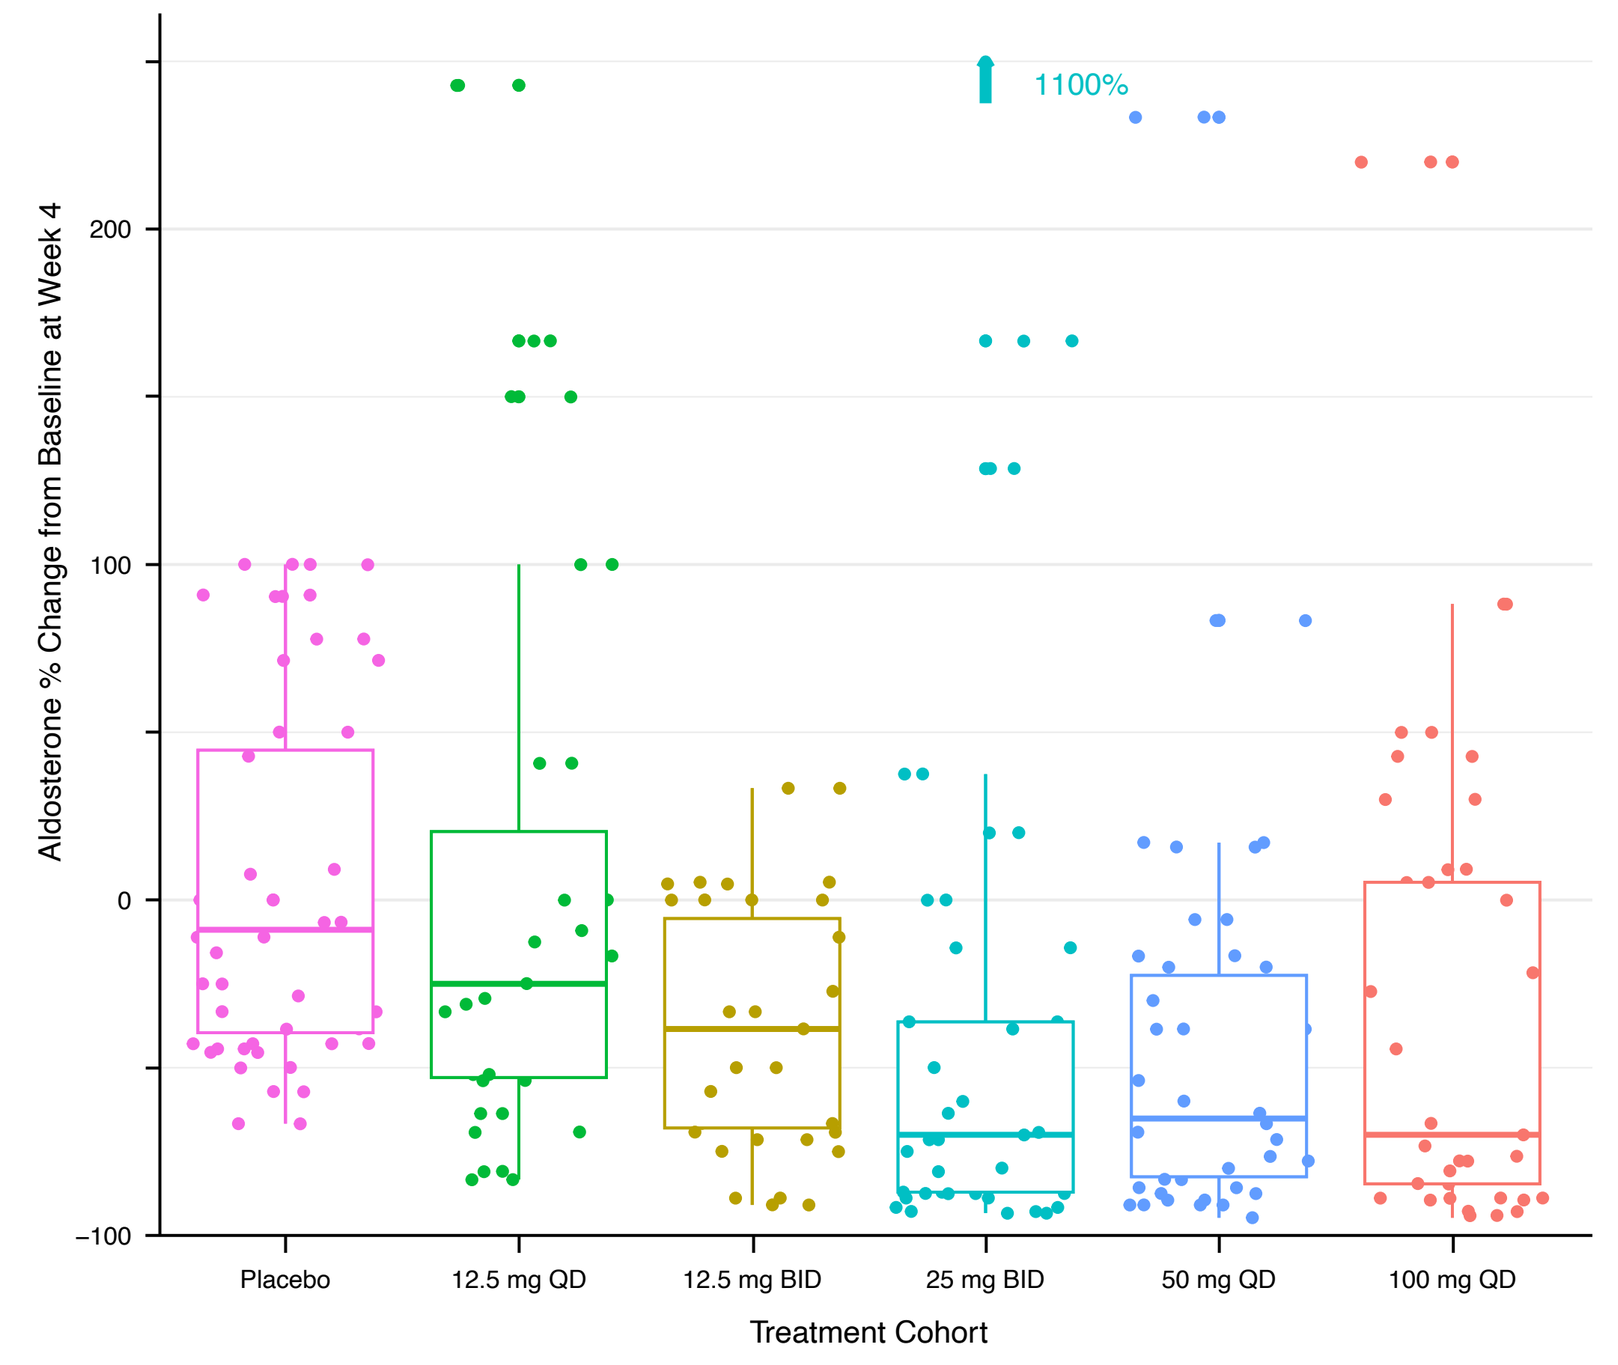
Aldosterone % Change from Baseline at Week 4
1100%
200
100
0
−100
Placebo
12.5 mg QD
12.5 mg BID
25 mg BID
50 mg QD
100 mg QD
Treatment Cohort

Daily dose (mg/24 h) in Part 1 cohort. BID, twice daily; QD, once daily.

Figure 4. Change in systolic BP at week 8

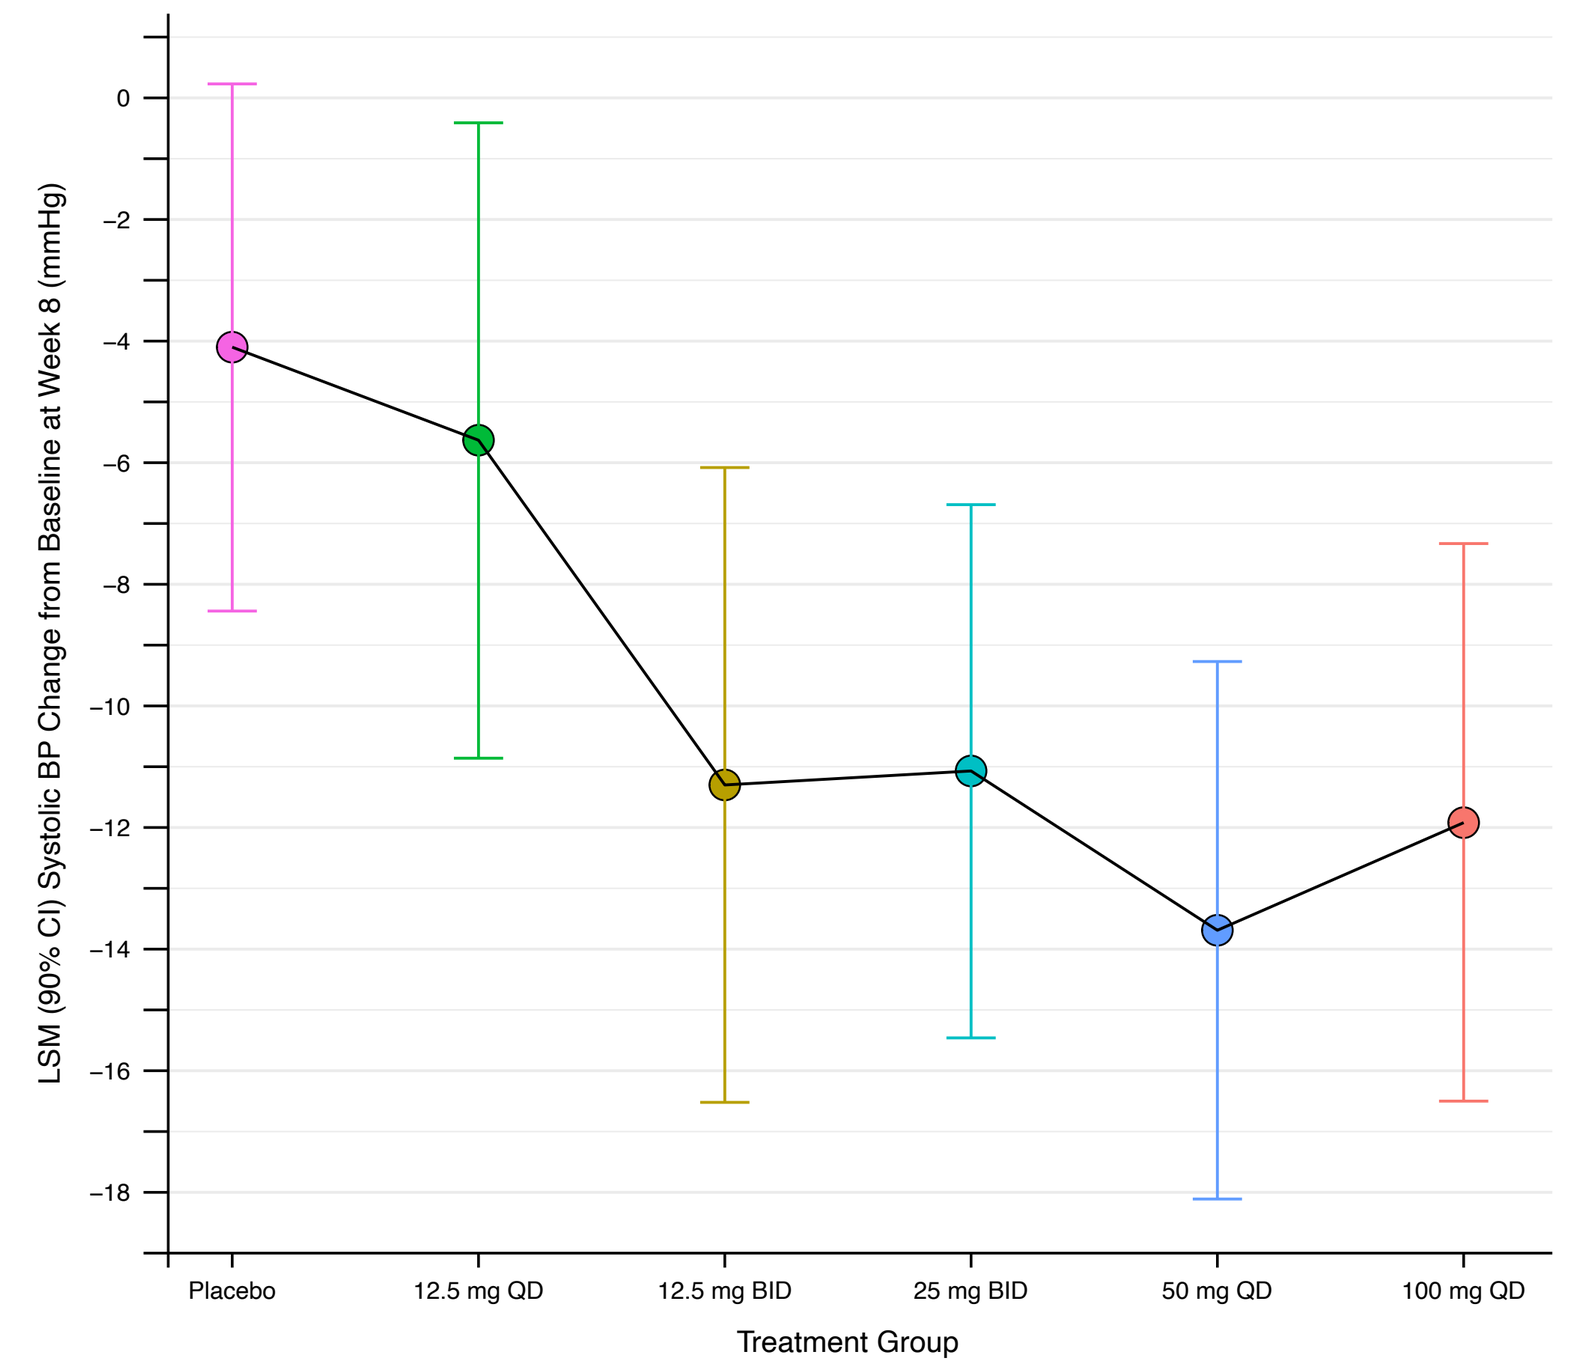
LSM (90% CI) Systolic BP Change from Baseline at Week 8 (mmHg)
0
−2
−4
−6
−8
−10
−12
−14
−16
−18
Placebo
12.5 mg QD
12.5 mg BID
25 mg BID
50 mg QD
100 mg QD
Treatment Group

Mixed model repeated measures and automated office blood pressure in Part 1 cohort. BID, twice daily; LSM, least square mean; QD, once daily.

Figure 5. Change in systolic BP at week 8 pooled across lorundrostat doses 25 mg BID, 50 mg QD, and 100 mg QD by (A) quartile groups and (B) frequency distribution

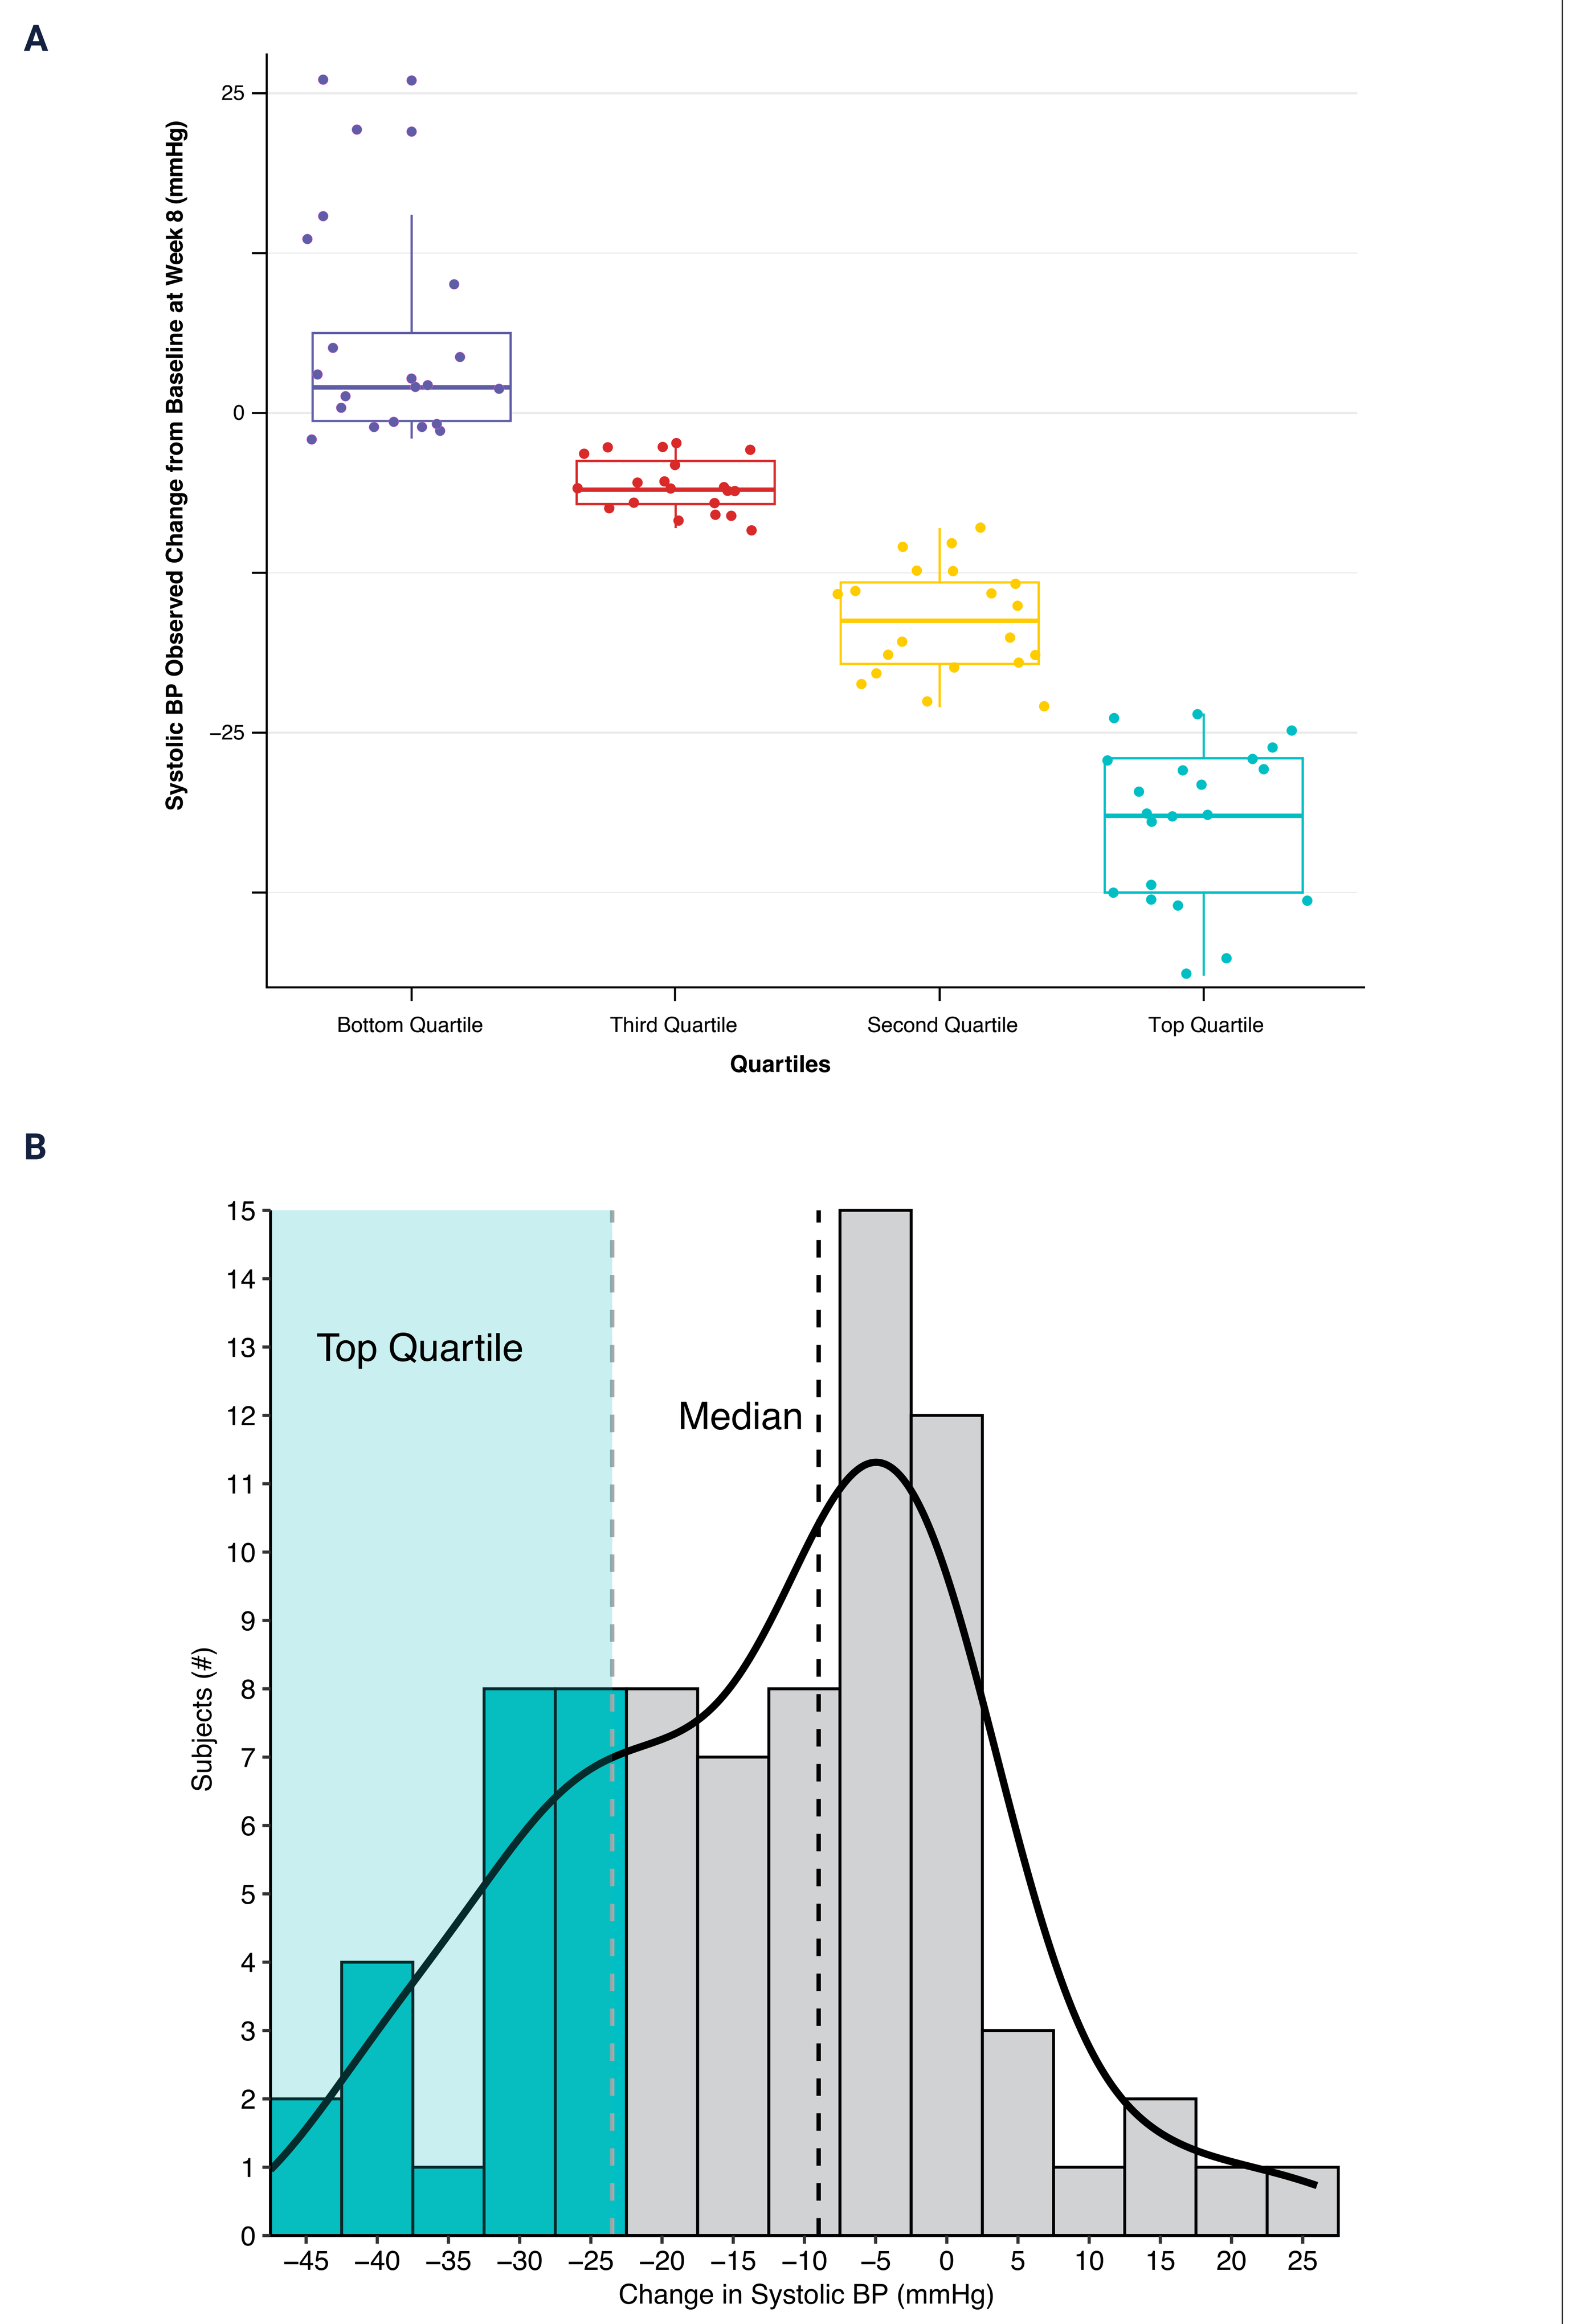
A
Systolic BP Observed Change from Baseline at Week 8 (mmHg)
25
0
−25
Bottom Quartile
Third Quartile
Second Quartile
Top Quartile
Quartiles
B
Subjects (#)
Top Quartile
Median
−45 −40 −35 −30 −25 −20 −15 −10 −5 0 5 10 15 20 25
Change in Systolic BP (mmHg)

Part 1 cohort (n=81) automated office blood pressure. The subjects in the top quartile (n=21) had a median (IQR) systolic BP reduction of -32 mmHg (-37.5 to -27.0) in response to lorundrostat. BID, twice daily; BP, blood pressure; IQR, interquartile range; QD, once daily.

Figure 6. Minimal relationship between baseline PRA and change in systolic BP at week 8

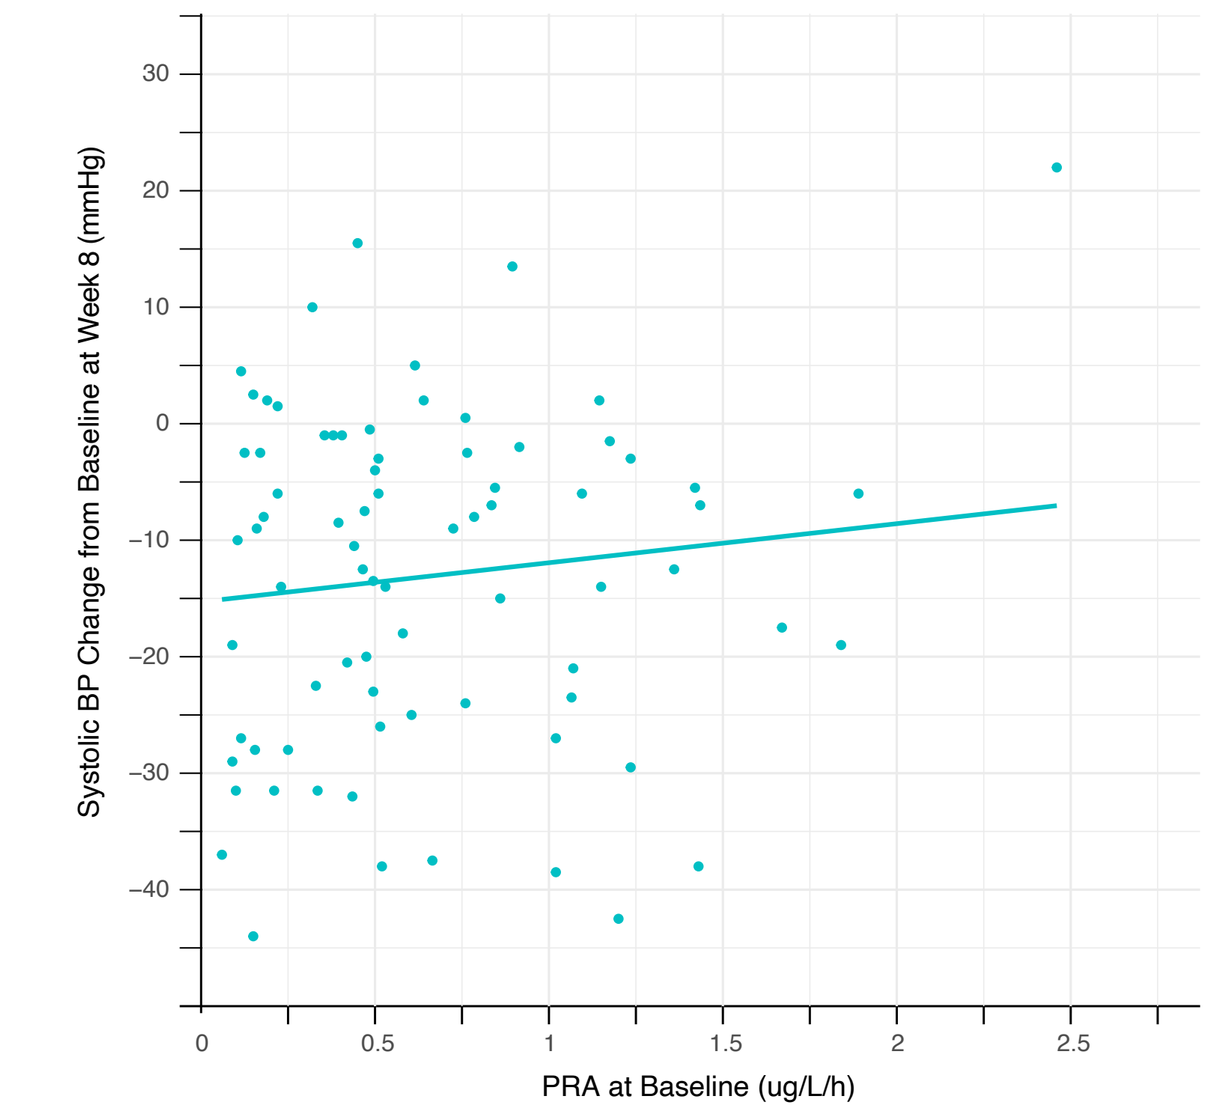
Systolic BP Change from Baseline at Week 8 (mmHg)
30
20
10
0
−10
−20
−30
−40
0
0.5
1
1.5
2
2.5
PRA at Baseline (ug/L/h)

Lorundrostat pooled dose 25 mg BID, 50 mg QD, and 100 mg QD in Part 1 cohort (n=81). Although Part 1 subjects were required to have PRA ≤1.0 at screening, some of these subjects had PRA >1.0 at the baseline assessment, and 4 subjects had high PRA values >3.0. Automated office blood pressure in full analysis set. BID, twice daily; PRA, plasma renin activity; QD, once daily.

Figure 7. Positive relationship between increased baseline BMI and reduction in systolic BP at week 8

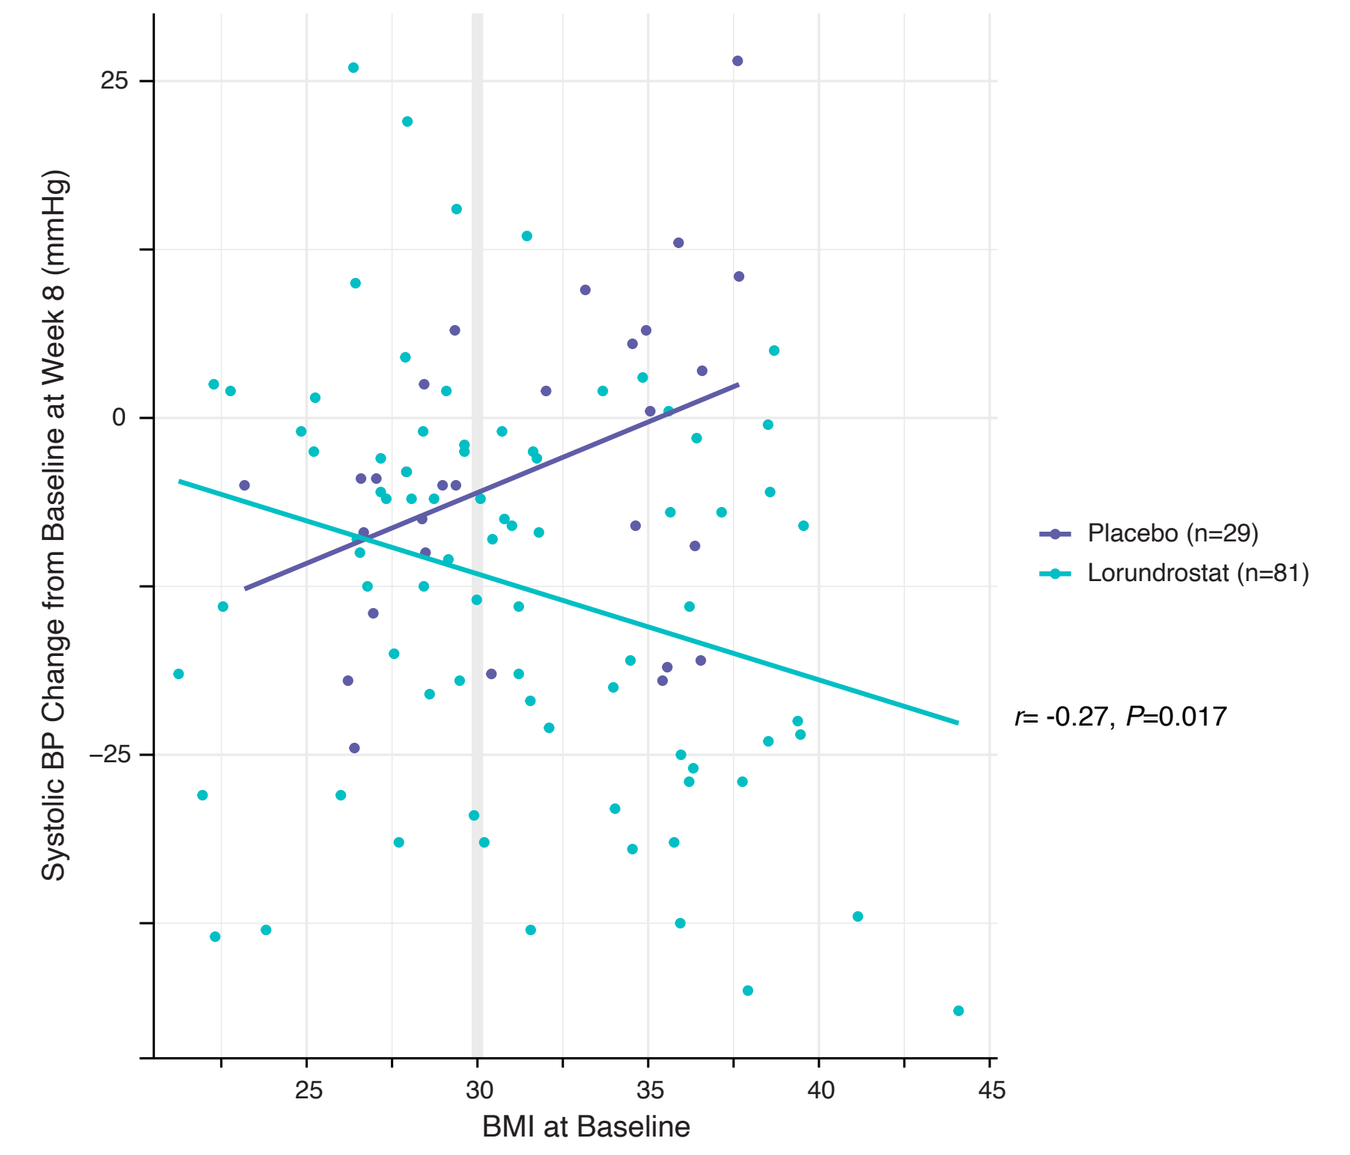
Systolic BP Change from Baseline at Week 8 (mmHg)
25
0
−25
25
30
35
40
45
BMI at Baseline
Placebo (n=29)
Lorundrostat (n=81)
r= -0.27, P=0.017

Lorundrostat pooled dose 25 mg BID, 50 mg QD, and 100 mg QD in Part 1 cohort. Automated office blood pressure in full analysis set. BID, twice daily; BMI, body mass index; BP, blood pressure; QD, once daily.

CONCLUSIONS

- **A population of adults with enhanced response to lorundrostat was identified. The enhanced response was associated with increasing BMI.**
- **An obesity-related, aldosterone-dependent hypertensive endotype indicates a need for a paradigm shift toward aldosterone targeted, precision therapy for hypertension management in these patients**

REFERENCES

1. Kawarazaki W, Fujita, T. *Am J Hypertens.* 2016;29(4):415-423.
2. Laffin, L, et al. *JAMA.* 2023;330(12):1140-1150.

ACKNOWLEDGMENTS

The study was sponsored by Mineralys Therapeutics, LLC. Medical writing and editing assistance were provided by Lynanne McGuire, PhD and Courtney Breuel, ELS of MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Mineralys Therapeutics, LLC.

DISCLOSURES

BC and DMR: Employee of Mineralys Therapeutics, LLC. **LL:** Cleveland Clinic, his employer, was a study site for the TARGET-HTN trial; C5Research, the academic research organization of the Cleveland Clinic, receives payment for services related to other Mineralys Therapeutics clinical trials; consultant to Medtronic, Lilly, and Crispr Therapeutics; received grants from AstraZeneca and stock options for LucidAct Health and Gordy Health. **NR:** Payment for contract research organization services provided by Cytel from Mineralys Therapeutics, LLC.

Presented at the American Society of Nephrology Kidney Week, November 2-5, 2023, Philadelphia, PA